Preliminary results of a phase 1, first-in-human, dose escalation study of the anti-CCR8 cytolytic antibody, CHS-114 (formerly SRF114) in patients with advanced solid tumors

Abstract #2664

Copies of this poster obtained through QR codes are for personal use only and may not be reproduced without written permission from ASCO® or the author of the poster.

Amita Patnaik*[1], Justin Call*[2], William McKean[2], Drew Rasco[1], Peter Oppelt[3], Varun N. Kapoor[4], Vinay Kumar[4], Narendiran Rajasekaran[4], Helen Tang[4], Vienna Reichert[4,5], Ricard Masia[4,5], Jessica Clement[5], Douglas Adkins[3]

*co-primary authors

[1] The START Center for Cancer Research, San Antonio, TX; [2] The START Center for Cancer Research, Salt Lake City, UT; [3] Washington University School of Medicine, Saint Louis, MO; [4] Coherus BioSciences, Redwood City, CA; [5] Surface Oncology, Cambridge, MA

INTRODUCTION

- FOXP3+ regulatory T cells (Tregs) play a crucial role in orchestrating immunosuppression within the TME, and their increased frequency within tumors correlates with poor clinical prognosis
- Past attempts at Treg depletion have lacked selectivity which resulted in autoimmune toxicity
- Chemokine receptor 8 (CCR8) is a G protein-coupled receptor (GPCR) that is highly upregulated by intratumoral Tregs compared to their peripheral counterparts and other immune cell types
- Anti-CCR8 mAb mediated depletion of intratumoral Tregs results in antitumor activity and T cell infiltration in a PD-1 resistant mouse model
- CHS-114 is an afucosylated mAb highly selective for CCR8 with no off-target binding
 - Afucosylation enhances cytolytic activity and promotes killing of intratumoral CCR8+ Tregs
 - Other CCR8-targeting mAbs currently in development lack selectivity and exhibit off-target binding
 - Minimal non-specific depletion of circulating Tregs mitigates risk of autoimmunity while maintaining efficacy
- CHS-114 has the potential to overcome Treg mediated immune suppression within the TME by recruiting T cells, turning cold tumors to hot, and enhancing antitumor immunity when combined with other IO agents, such as toripalimab
- **A Phase 1, first-in-human (FIH) clinical trial was initiated to evaluate CHS-114 in patients with advanced solid tumors and head and neck squamous cell carcinoma (HNSCC)**

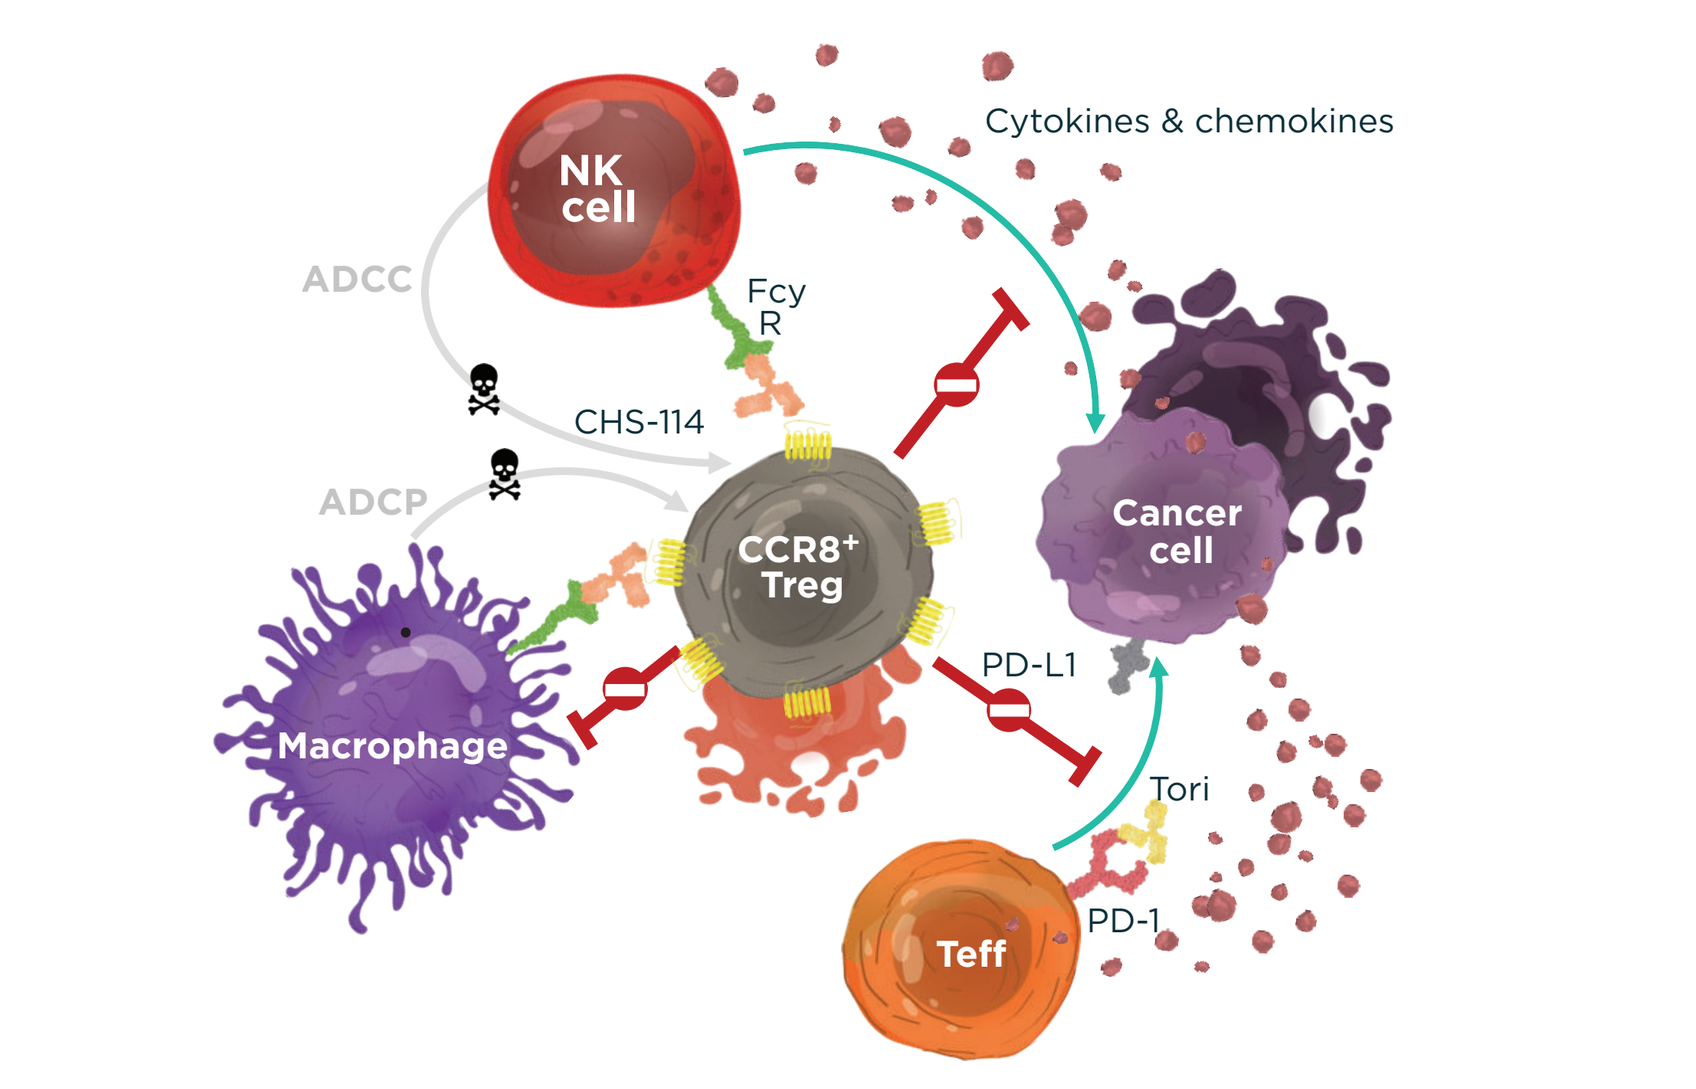
Cytokines & chemokines
NK cell
ADCC
Fcγ R
CHS-114
ADCP
CCR8+ Treg
Cancer cell
Macrophage
PD-L1
Tori
PD-1
Teff

CHS-114 Selectively Binds CCR8, A GPCR Preferentially Expressed on Tumor Resident Treg Cells

Selected Antibodies	Number of Non-CCR8 Targets Identified
CHS-114	0
mAb2	1
mAb3	8
mAb4	15
mAb5	20
Comparator Antibodies	**Non-CCR8, "Off Target" Binding**
Comparator 1	ANGPTL7
Comparator 2	J chain
Comparator 3	SEMA4B

GPCRs are challenging antibody targets due to limited surface protein accessibility; as a result GPCR targeted mAbs often show off target binding. A screen of 5,528 proteins using cell microarray technology was performed for CHS-114 and 3 other clinical stage CCR8 mAb (antibodies derived from patent sequences).

CCR8 Cell Depletion Enhances PD-1 Antitumor Immunity and Increases Tumor Infiltrating CD8+ T Cells.

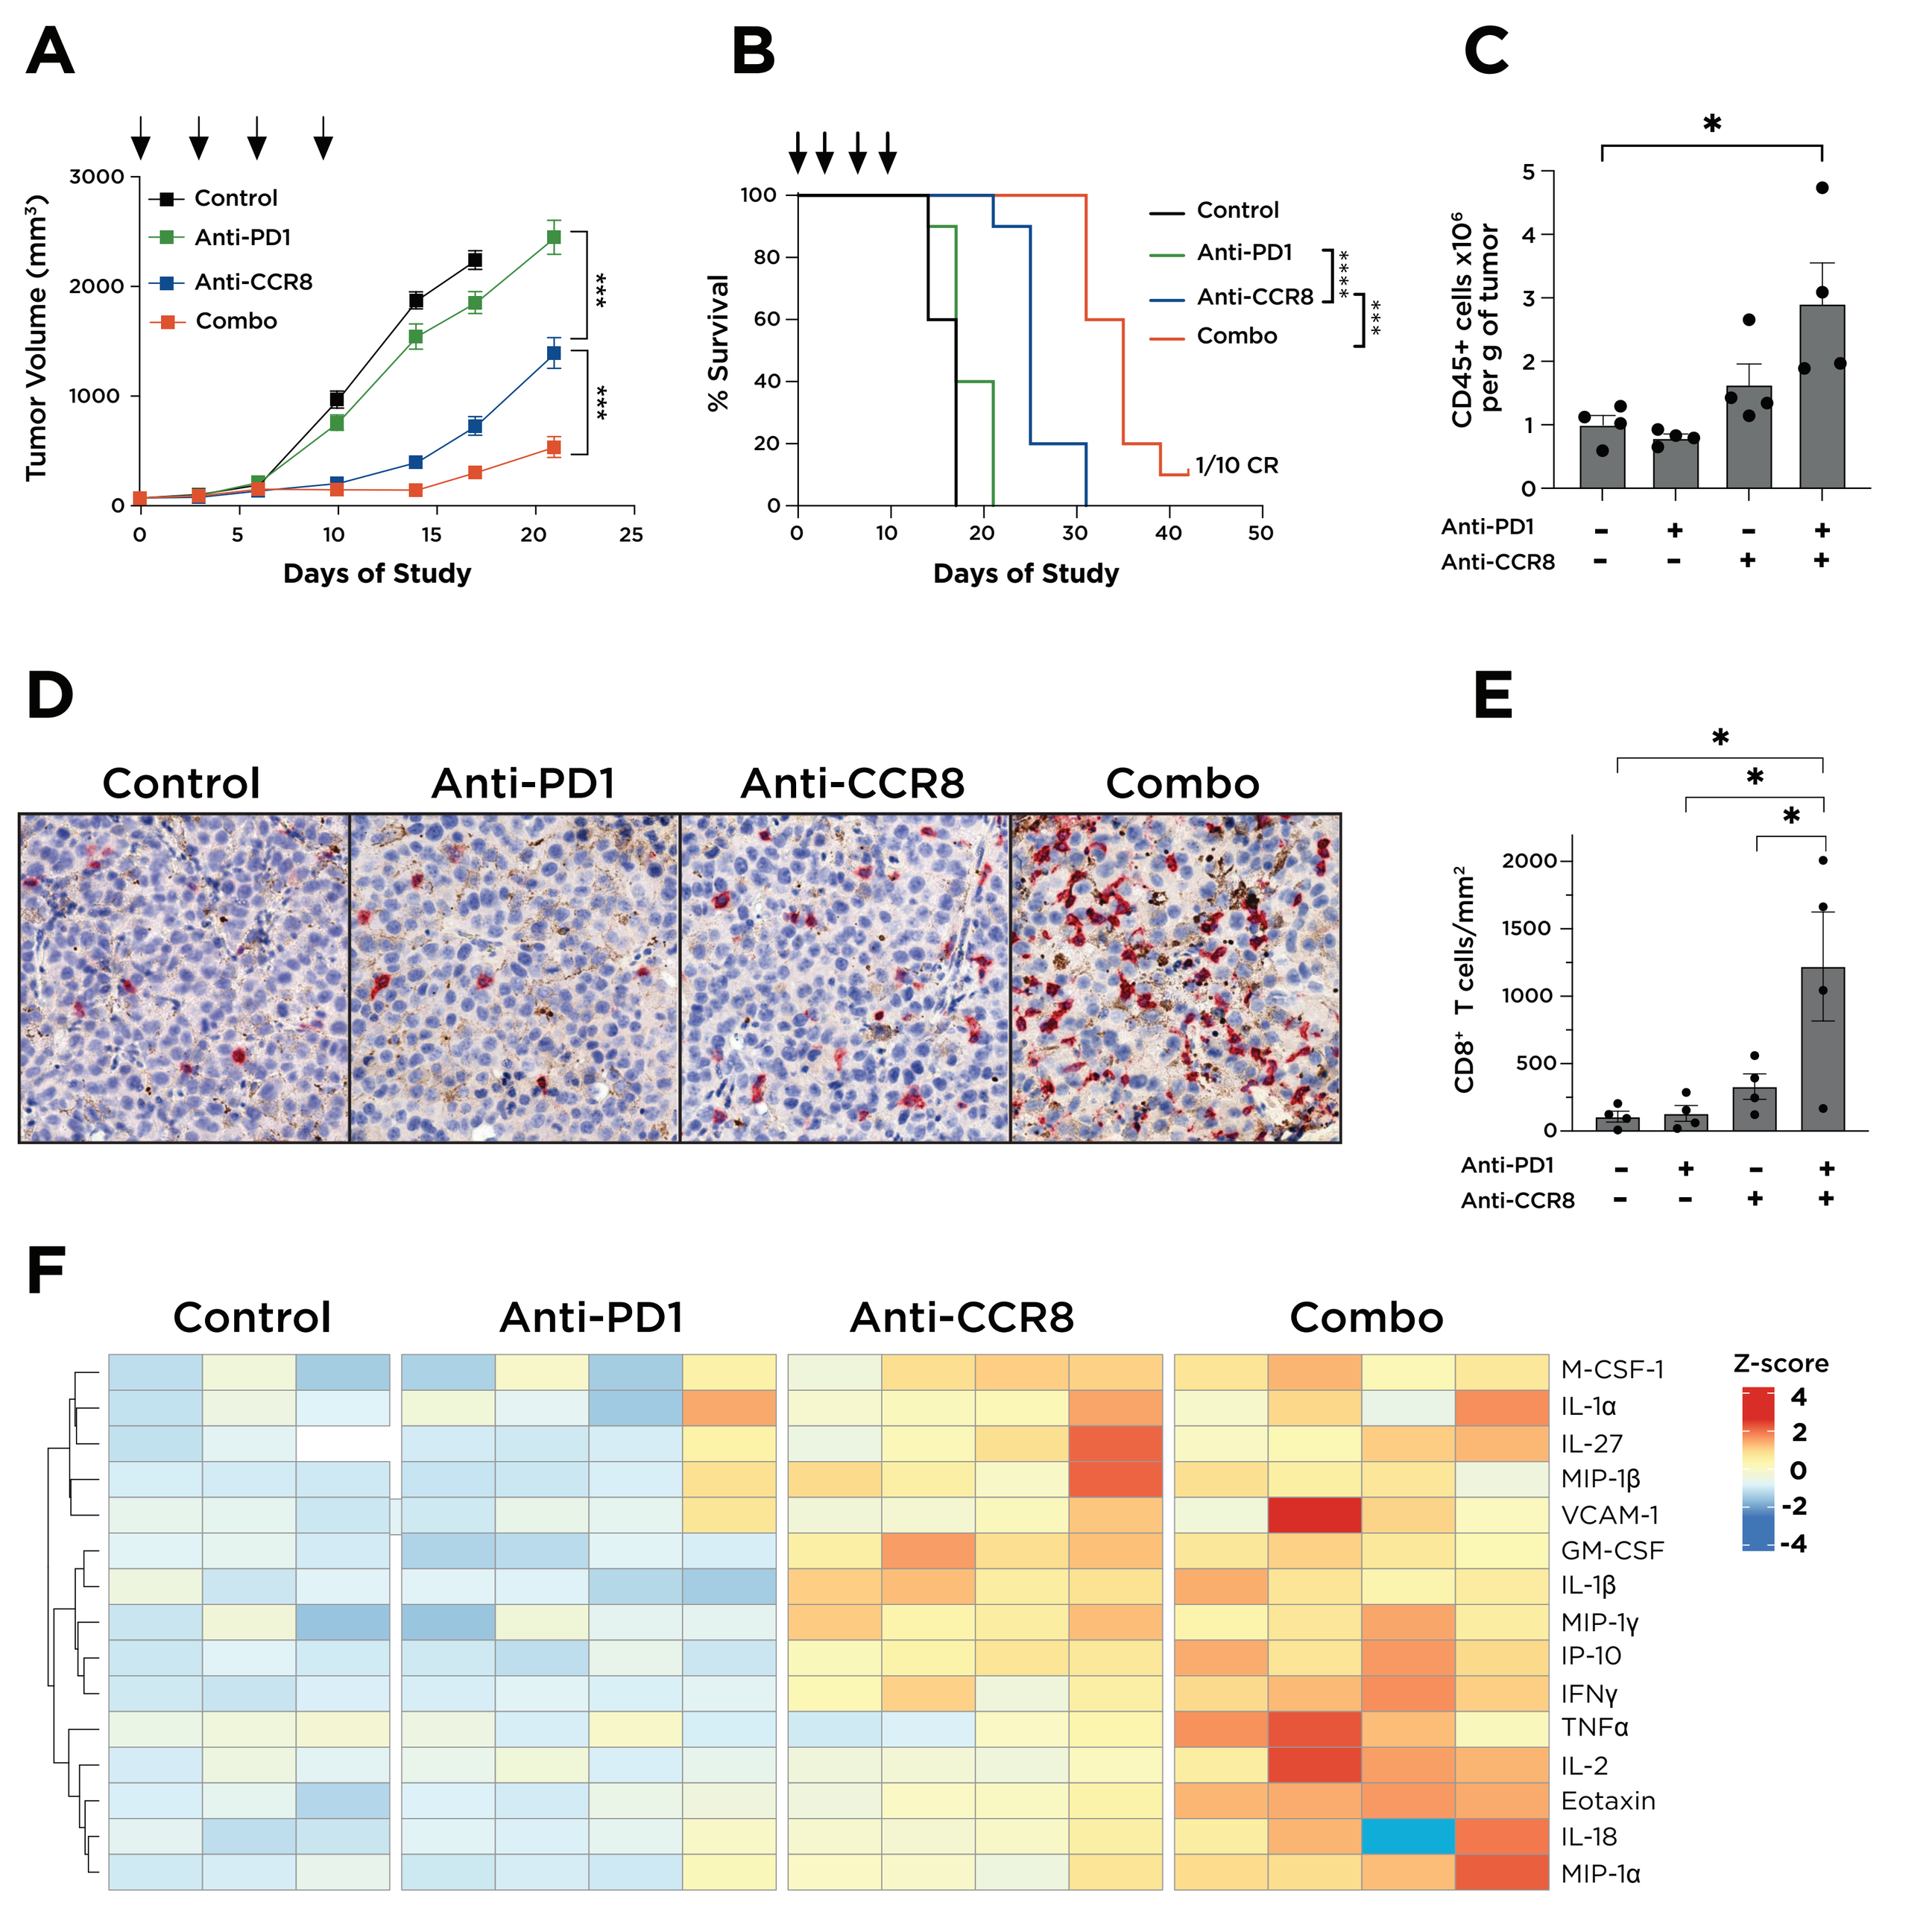
A
Tumor Volume (mm³)
Control
Anti-PD1
Anti-CCR8
Combo
Days of Study
B
% Survival
1/10 CR
Days of Study
C
CD45+ cells x10⁶ per g of tumor
Anti-PD1
Anti-CCR8
D
Control
Anti-PD1
Anti-CCR8
Combo
E
CD8+ T cells/mm²
F
Control
Anti-PD1
Anti-CCR8
Combo
Z-score
M-CSF-1
IL-1α
IL-27
MIP-1β
VCAM-1
GM-CSF
IL-1β
MIP-1γ
IP-10
IFNγ
TNFα
IL-2
Eotaxin
IL-18
MIP-1α

1×10^5 B16F10 cells were injected to the flank of C57BL/6 mice subcutaneously. Mice were randomized to four groups and dosing with isotype control antibodies, anti-CCR8 (mouse IgG2a, 50 µg/mouse), anti-PD-1 (rat IgG2a, 200 µg/mouse), or combo treatments on day 6 post tumor implantation (n=10/group).

A) Anti-tumor activity (tumor growth inhibition assessment). **B)** Survival curves. **C)** Tumor cytometric analyses and average frequency, respectively, of TILs on Study Day 9. Total number of CD45+ cells per gram of tumor was measured by normalizing the total CD45+ cell count to tumor weight (n=4) **(D)** Representative IHC images of tumor infiltrating CD8+ T cells (red chromogen, Study Day 9). **E)** Quantification of CD8+ T cells using image analysis on digitally scanned slides (n=4). **F)** Production of pro-inflammatory cytokines and chemokines in B16F10 tumors. Cytokines and chemokines were detected by Luminex assay in snap-frozen and lysed tumors Study Day 8 (n=3-4). Statistical analysis was performed using log-rank test **(B)** or one-way ANOVA **(A, C, E)**. $^*p<0.05$, $^{**}p<0.01$, $^{***}p<0.001$, $^{****}p<0.0001$.

Reference: Panduro, Marisella, et al. "Depletion of CCR8+ tumor Treg cells with SRF114 or anti-CCR8 therapy promotes robust antitumor activity and reshapes the tumor microenvironment toward a more pro-inflammatory milieu." *Cancer Research* 83.7_Supplement (2023): 5125-5125.

METHODS

- Phase 1, FIH, open-label, single-agent and combination therapy dose escalation (NCT05635643)
- Stage 1 (CHS-114 single-agent dose escalation) employed the Bayesian optimal interval (BOIN) design including accelerated titration and 3+3 run-in
 - Stage 1a enrolled patients with advanced solid tumors who received ≥1 standard treatment
 - Stage 1b, is enrolling an additional 5 patients with advanced/metastatic HNSCC at each of two dose levels (DLs); patients must be willing to undergo pre- and on-treatment biopsies
- Stage 2 (CHS-114 + toripalimab combination dose escalation) is enrolling patients with advanced/metastatic HNSCC and employ a standard 3+3 design
- CHS-114 is administered intravenously (IV) on day 1 of each Q3W cycle; in Stage 2, CHS-114 will be administered in combination with toripalimab 240mg Q3W
- Dose-limiting toxicities (DLTs) were evaluated during Cycle 1 (21 days) using NCI-CTCAE criteria (v5.0 or higher)

SRF114-101 Study Design

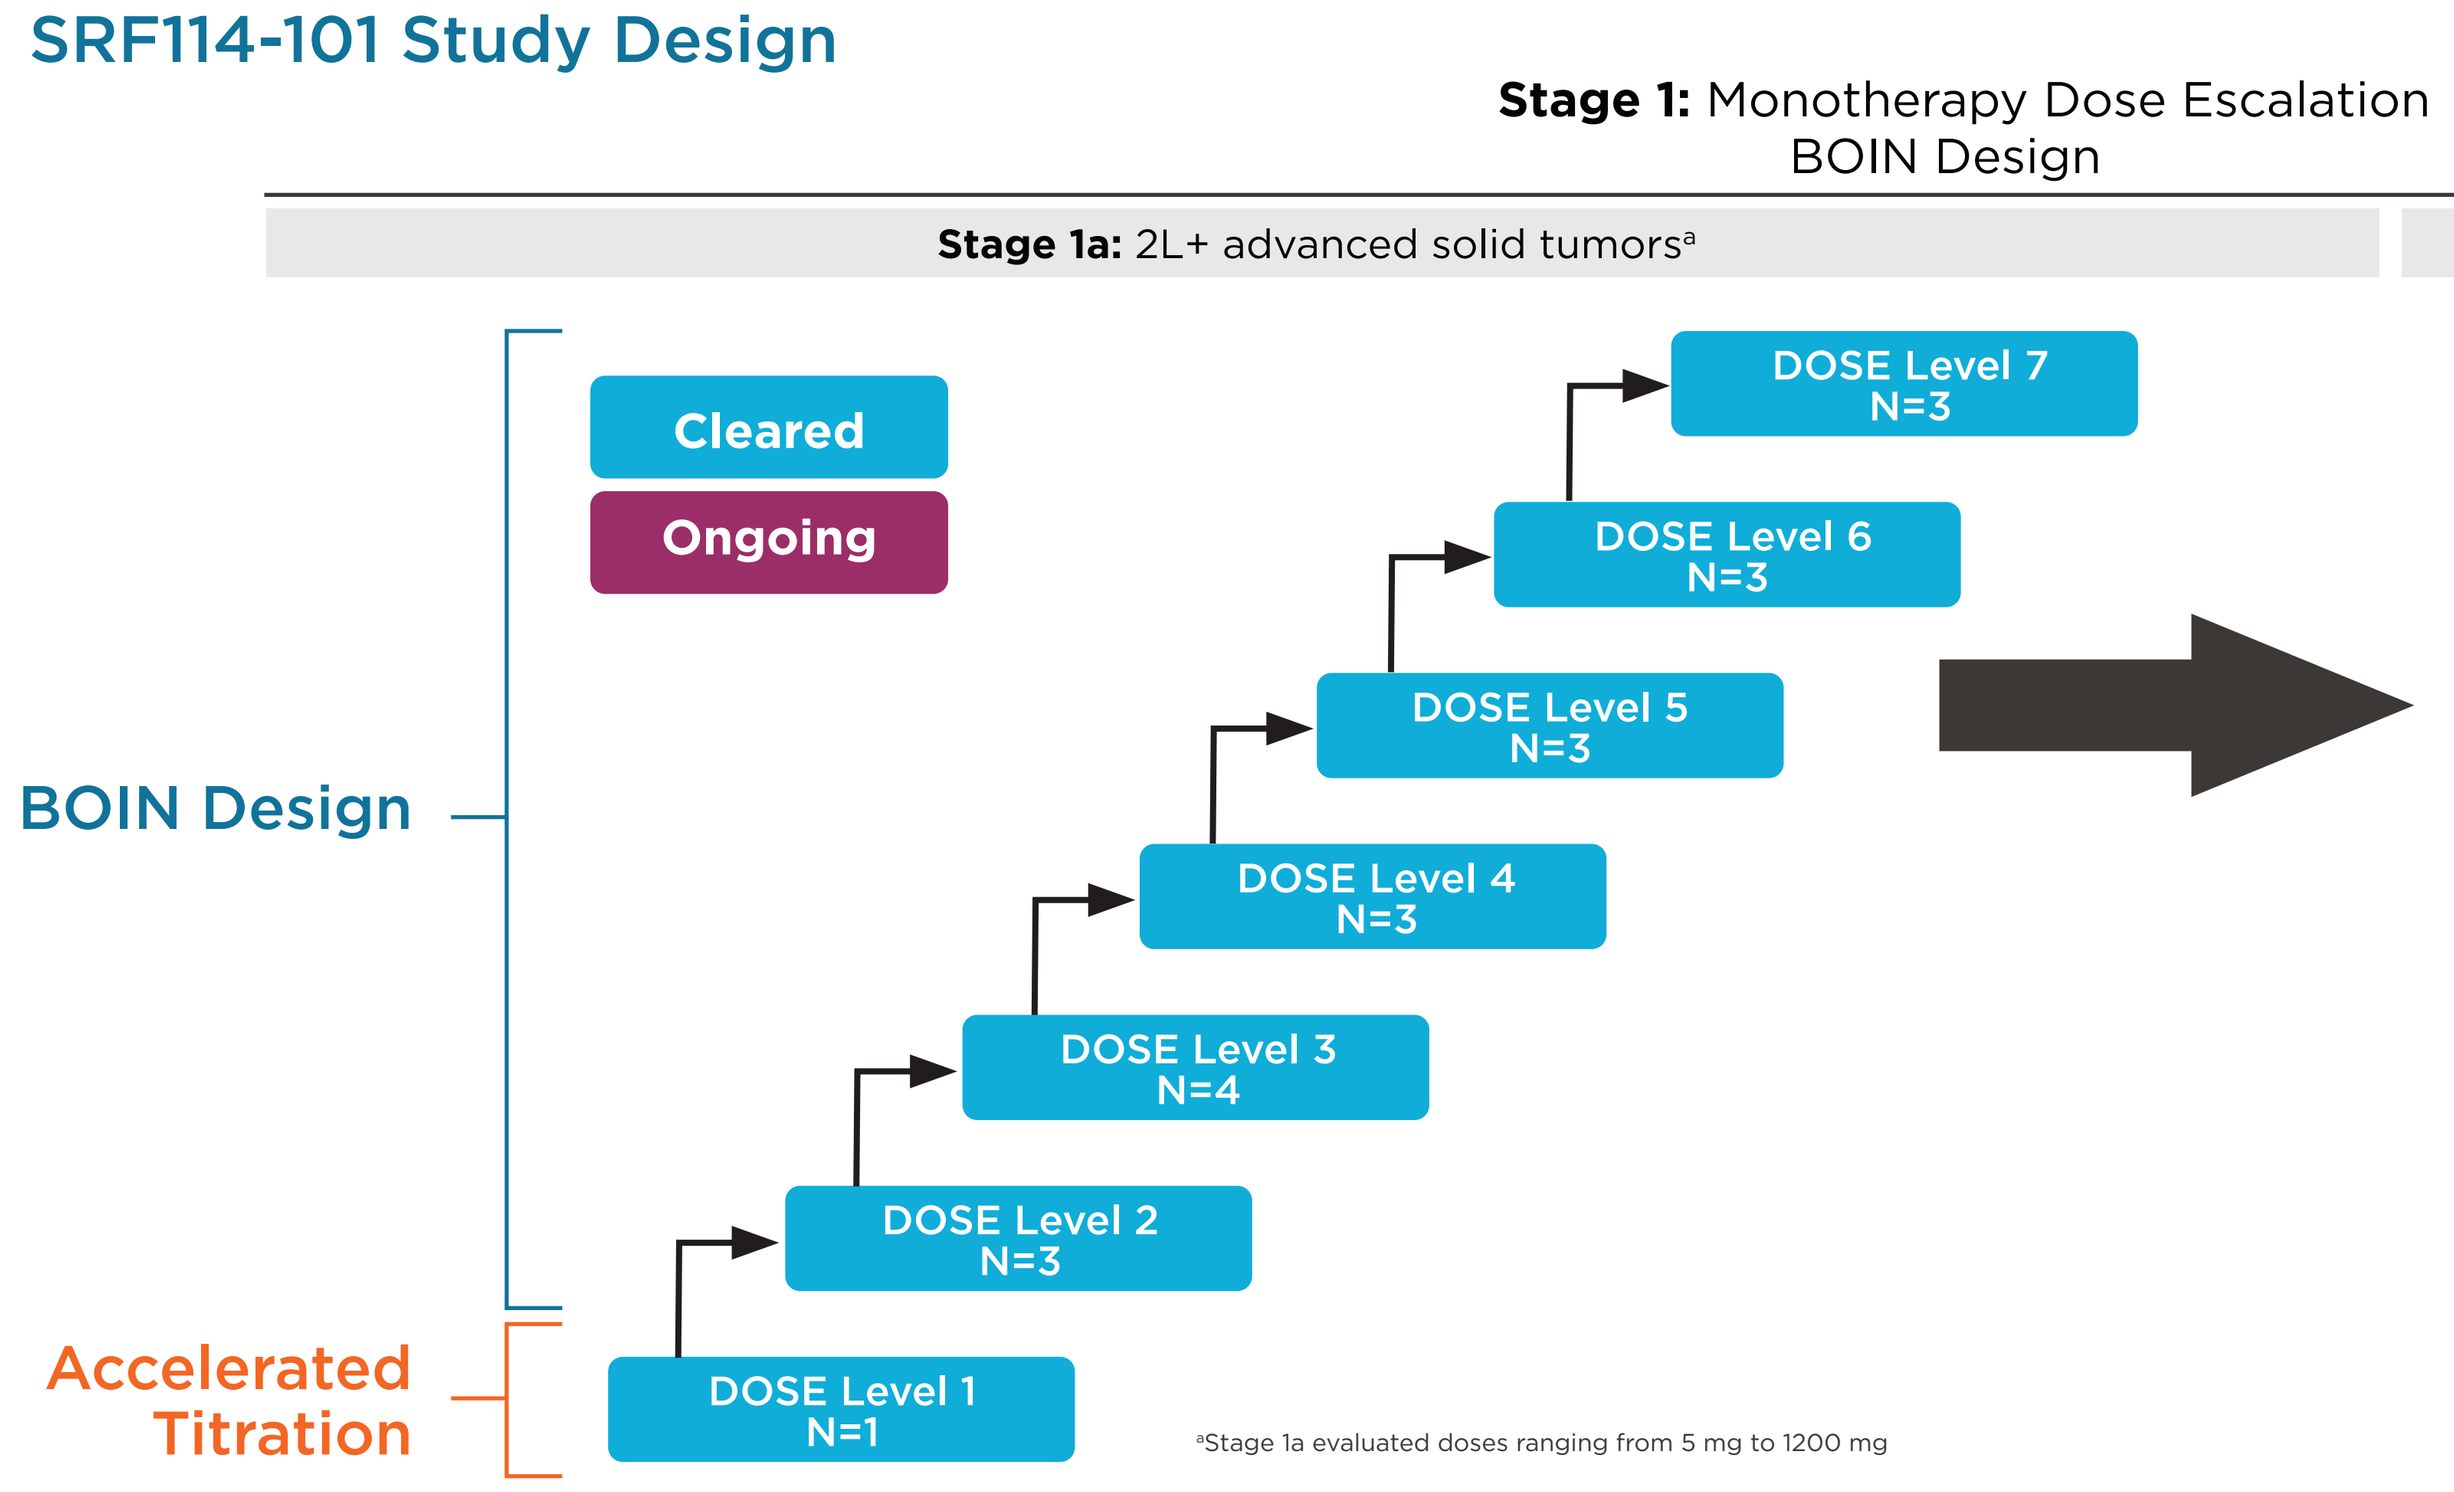
Stage 1: Monotherapy Dose Escalation
BOIN Design
Stage 1a: 2L+ advanced solid tumors^a
Stage 1b: 2L+ HNSCC
Cleared
Ongoing
BOIN Design
Accelerated Titration
DOSE Level 1 N=1
DOSE Level 2 N=3
DOSE Level 3 N=4
DOSE Level 4 N=3
DOSE Level 5 N=3
DOSE Level 6 N=3
DOSE Level 7 N=3
^a Stage 1a evaluated doses ranging from 5 mg to 1200 mg

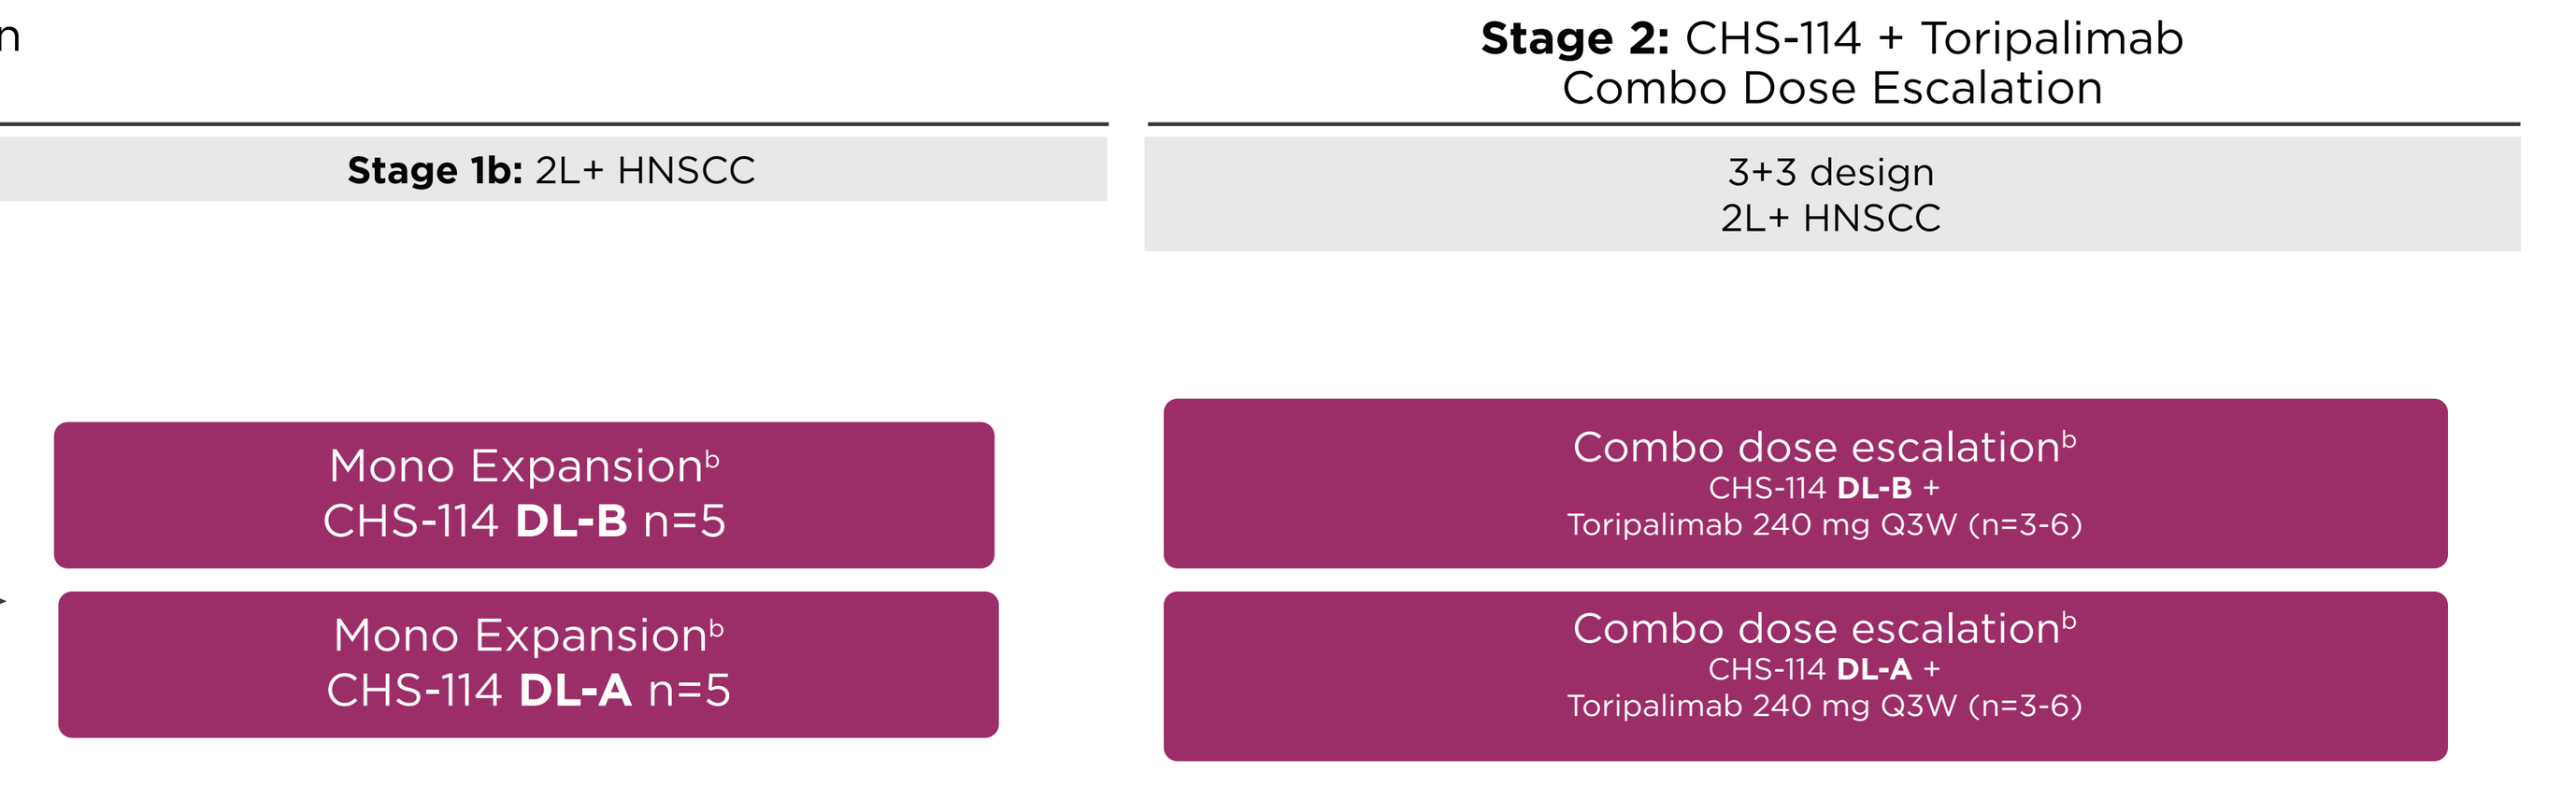
Stage 2: CHS-114 + Toripalimab
Combo Dose Escalation
3+3 design
2L+ HNSCC
Mono Expansion^b
CHS-114 DL-B n=5
Mono Expansion^b
CHS-114 DL-A n=5
Combo dose escalation^b
CHS-114 DL-B +
Toripalimab 240 mg Q3W (n=3-6)
Combo dose escalation^b
CHS-114 DL-A +
Toripalimab 240 mg Q3W (n=3-6)

- Primary endpoints: rate of DLTs and TEAES, with overarching objective of determining two recommended doses for expansion (RDE)
- Key secondary endpoints: objective response rate (ORR) based on Investigator review per RECIST v1.1, pharmacokinetics, pharmacodynamic assessments (changes in FOXP3 expression within tumor tissue – Stage 1b)
- Exploratory pharmacodynamic endpoint: Changes in frequency of CCR8-expressing immune cell subsets in the periphery

^b May be enrolled concurrently
2L: second-line; HNSCC: head and neck squamous cell carcinoma.

RESULTS

Dose Escalation Baseline Characteristics

Demographics n (%)		(n=20)
Age	Median years (Range)	67 (47, 84)
Gender	Female	10 (50)
	Male	10 (50)
Primary Tumor Type	Colorectal	4 (20)
	Endometrial	2 (10)
	Head and Neck Squamous Cell Carcinoma (HNSCC)	2 (10)
	Kidney	1 (5)
	Melanoma	1 (5)
	Non-Small Cell Lung	2 (10)
	Pancreatic	3 (15)
	Other*	5 (25)

Baseline Characteristics n (%)		(n=20)
ECOG PS	0	6 (30)
	1	14 (70)
Median Time Since Initial Diagnosis, Months (range)		47 (11, 257)
Lines of Prior Systemic Therapy	0	0
	1-2	5 (25)
	3-4	6 (30)
	≥5	9 (45)
PD-L1 Expression	Positive	6 (30)
	Negative	5 (25)
	Not Done	9 (45)

ECOG PS, Eastern Cooperative Oncology Group Performance Status; PD-L1, programmed death ligand 1
*Other tumor types include biliary tract (n=1), esthesioneuroblastoma (n=1), ovarian (n=3), and rectal (n=1).

Safety Summary

AE Summary	N=20
Treatment emergent adverse event (TEAE), n (%)	19 (95)
CHS-114-related AE, n (%)	8 (40)
Grade ≥3 TEAE, n (%)	7 (35)
Grade ≥3 treatment-related AE, n (%)	0
Serious Treatment Emergent Adverse Event (TESAE), n (%)	6 (30)
Treatment-related SAE, n (%)	1 (5)
TEAE leading to CHS-114 discontinuation, n (%)	1 (5)
Treatment-related AE leading to CHS-114 discontinuation, n (%)	0
TEAE leading to death, n (%)	1 (5)
Treatment-related AE leading to death, n (%)	0

Data cut as of 16 April 2024; subject to change

- No DLTs observed to date, across all dose levels tested
- Treatment-related TEAEs were generally low grade, with the most frequent being diarrhea, nausea, chills and pyrexia, each reported in 2 patients
- 1 patient experienced a treatment-related SAE of Grade 2 colitis

TEAEs and Treatment-Related TEAEs

Majority of TEAEs Consistent With Advanced Disease; Low Rate of High Grade TEAEs

TEAEs and Treatment-Related TEAEs Occurring in ≥ 10% of Patients (N=20)

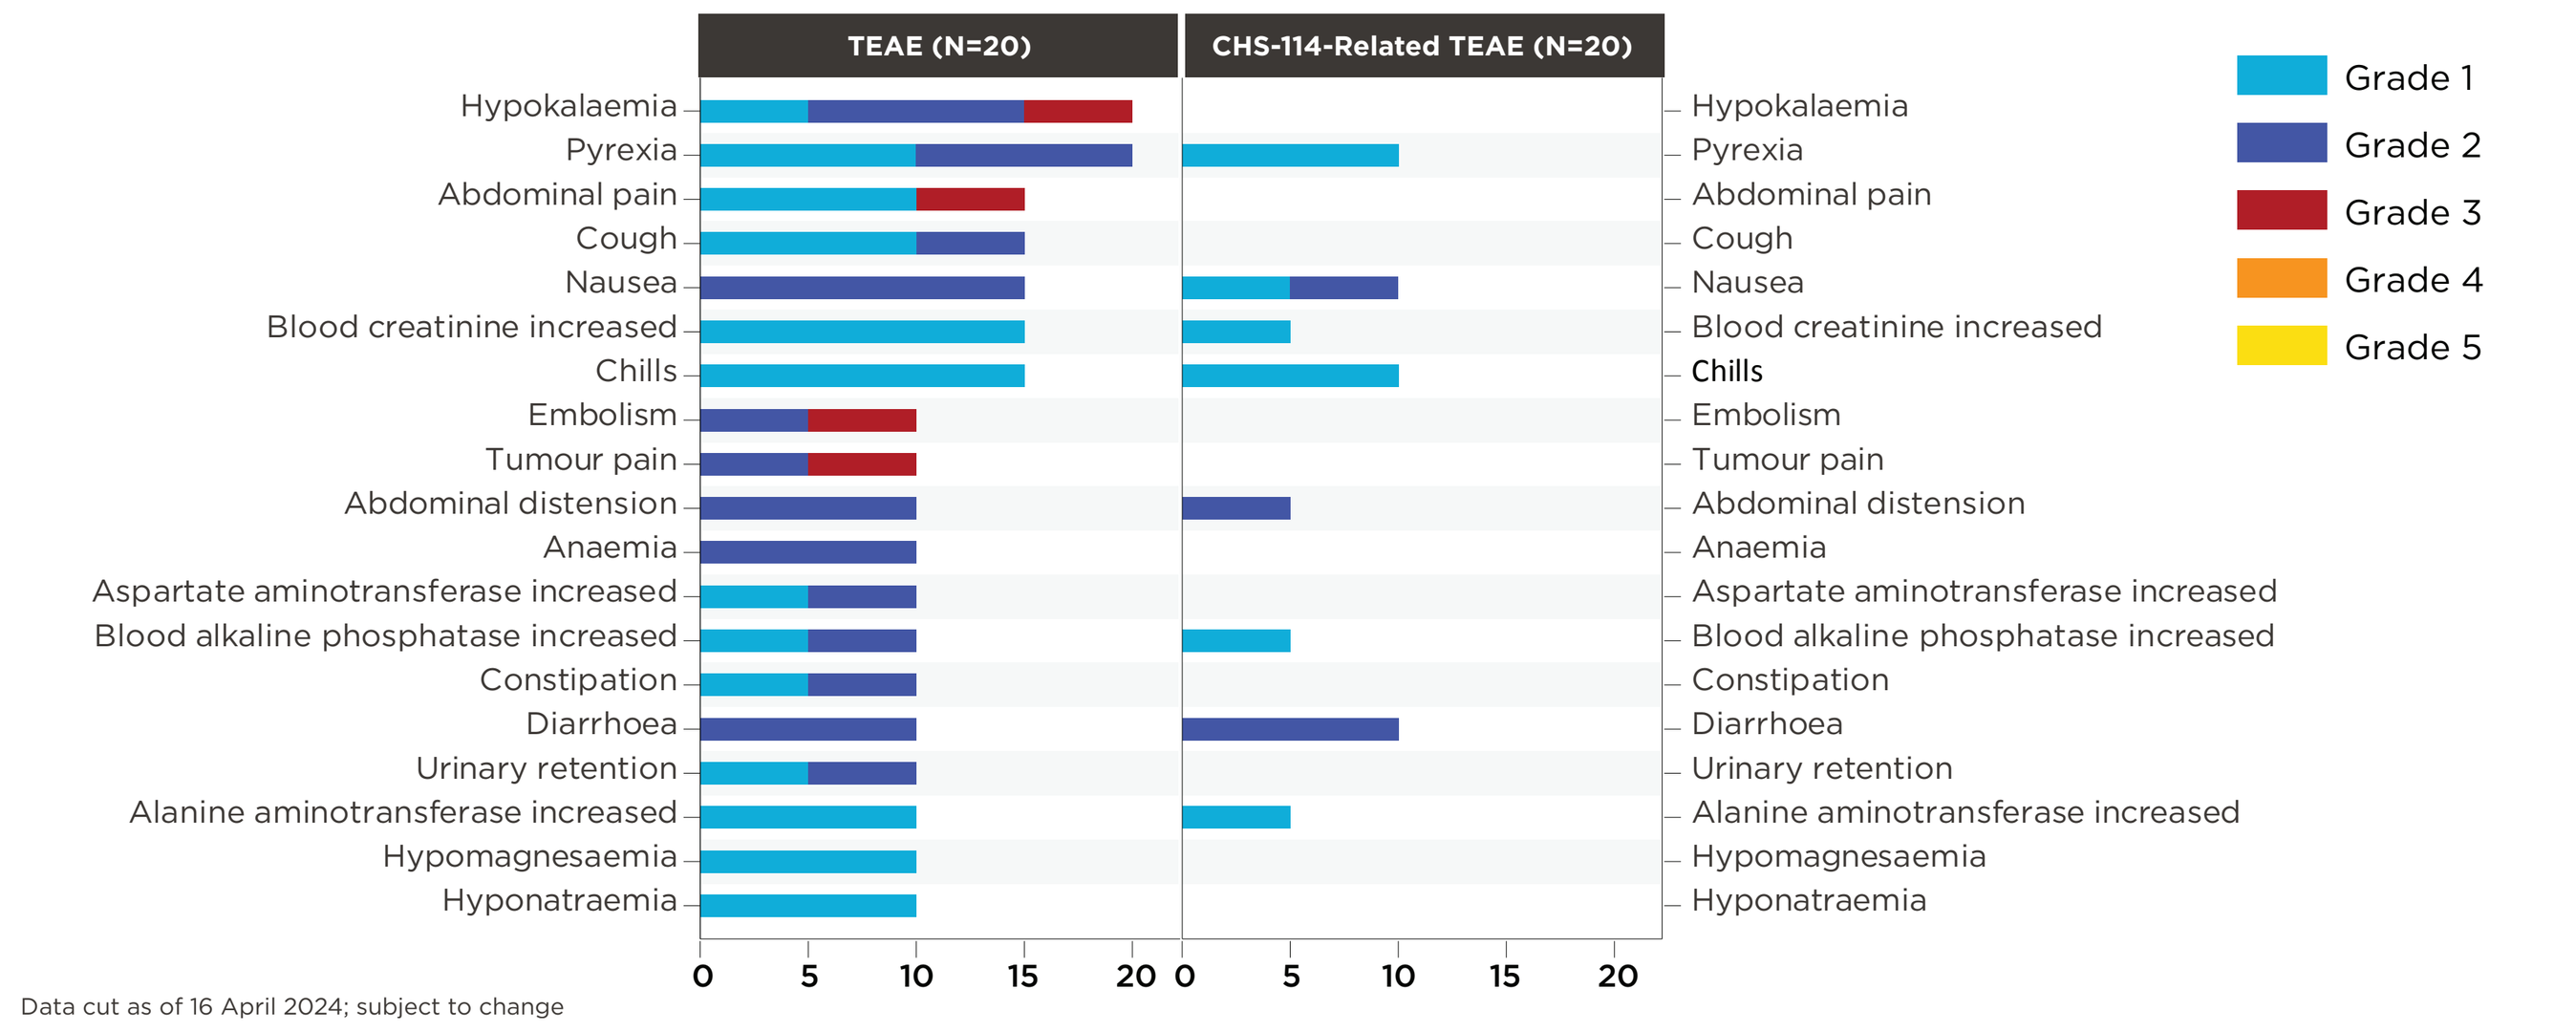
TEAE (N=20)
CHS-114-Related TEAE (N=20)
Grade 1
Grade 2
Grade 3
Grade 4
Grade 5
Hypokalaemia
Pyrexia
Abdominal pain
Cough
Nausea
Blood creatinine increased
Chills
Embolism
Tumour pain
Abdominal distension
Anaemia
Aspartate aminotransferase increased
Blood alkaline phosphatase increased
Constipation
Diarrhoea
Urinary retention
Alanine aminotransferase increased
Hypomagnesaemia
Hyponatraemia

Data cut as of 16 April 2024; subject to change

Response Summary based on Investigator Assessment per RECIST v1.1

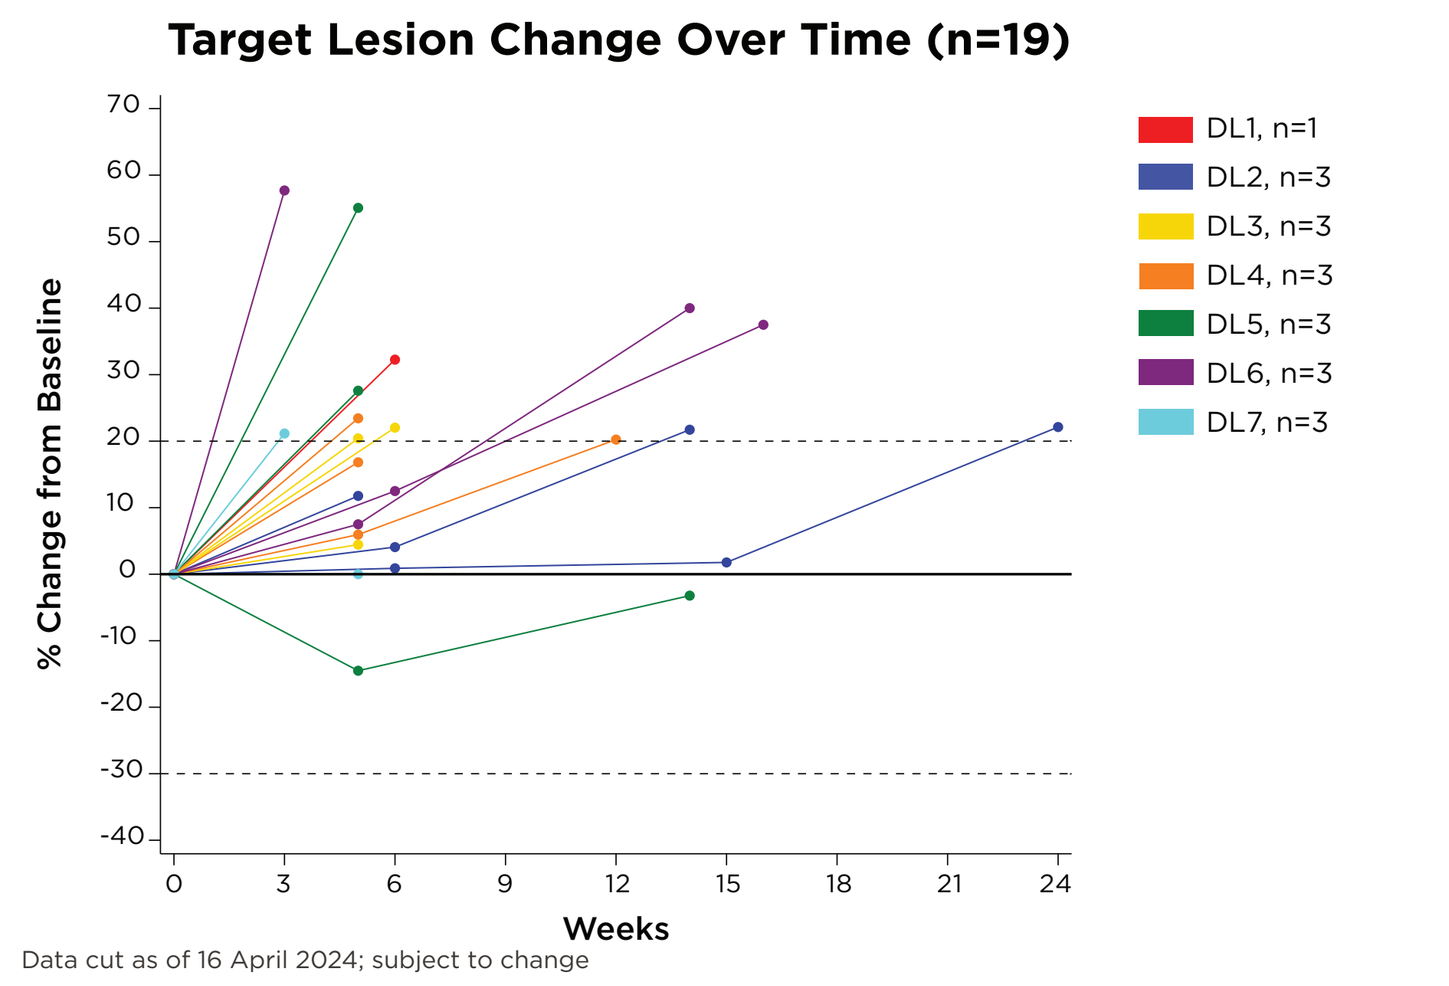
Target Lesion Change Over Time (n=19)
% Change from Baseline
Weeks
DL1, n=1
DL2, n=3
DL3, n=3
DL4, n=3
DL5, n=3
DL6, n=3
DL7, n=3

Data cut as of 16 April 2024; subject to change

Best overall response, n (%)	Response evaluable (n=19)
Complete response	0
Partial response	0
Stable disease	9 (47.4)
Progressive disease	10 (52.6)

Disease assessments performed every 9 weeks

Pharmacology: Sustained Peripheral CCR8+ Treg Depletion and Pharmacokinetic Profile

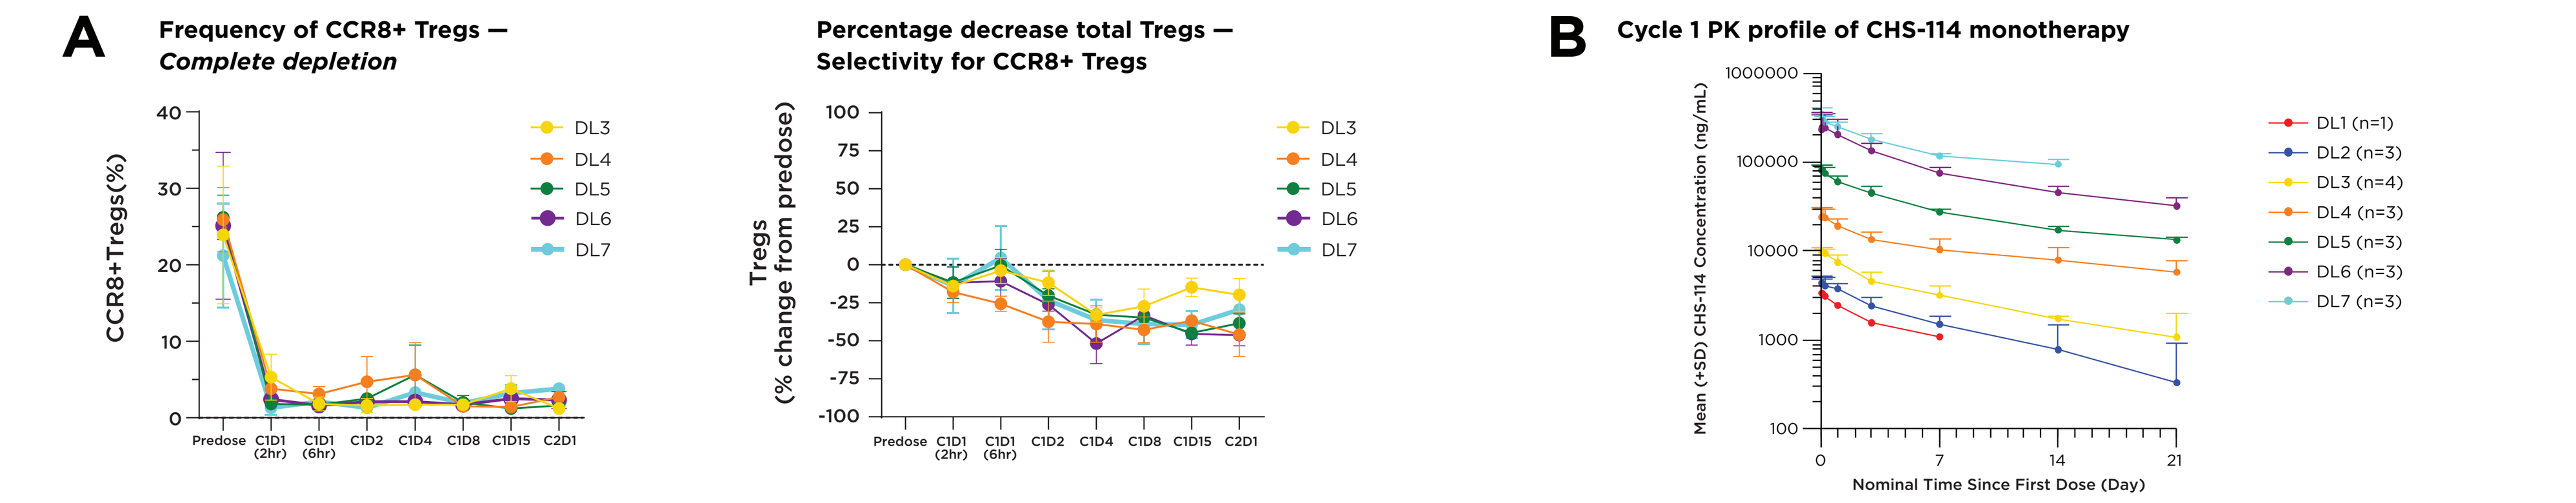
A
Frequency of CCR8+ Tregs — Complete depletion
CCR8+Tregs(%)
Percentage decrease total Tregs — Selectivity for CCR8+ Tregs
Tregs (% change from predose)
DL3
DL4
DL5
DL6
DL7
Predose
C1D1 (2hr)
C1D1 (6hr)
C1D2
C1D4
C1D8
C1D15
C2D1
B
Cycle 1 PK profile of CHS-114 monotherapy
Mean (+SD) CHS-114 Concentration (ng/mL)
Nominal Time Since First Dose (Day)
DL1 (n=1)
DL2 (n=3)
DL3 (n=4)
DL4 (n=3)
DL5 (n=3)
DL6 (n=3)
DL7 (n=3)

A) CHS-114 selectively depletes CCR8+ Tregs. Total frequency of CCR8+ Tregs from baseline (left) and percent decrease of total Tregs (right) was measured in peripheral blood mononuclear cells (PBMC) by a flow-cytometry assay at DL3-DL7. CCR8+ Treg depletion was stable through cycle 1, with > 85% of CCR8+ Tregs being depleted for all dose levels tested, confirming the proof of mechanism. Additionally, depletion was observed at DL3 (and higher doses), which was lower than predicted dose from *in vitro* modeling. Furthermore, CHS-114 treatment led to a decrease in subset of total Tregs, while preserving broader Treg population, confirming the specificity of CHS-114 for CCR8+ Tregs. Tregs were defined as CD127low CD25high cells within the CD3+ CD4+ T cell population. Data representative of 3 patients per dose level (n=2-3 samples per timepoint). Error bars = SEM.

B) CHS-114 Mean (+SD) serum concentration (Semi-Log) - time profile after single dose of CHS-114 (cycle 1). CHS-114 PK exposure increases with dose. Dose proportionality is observed at all dose levels. PK elimination is linear over all dosing levels with a half-life of approximately 10 days (range: 9-17 days). The PK datacut date was March 28, 2024.

CONCLUSIONS

- CCR8 is a GPCR that shows preferential expression on tumor resident Tregs and has promise as a drug target for selectively targeting immune suppression in the tumor microenvironment
- CHS-114 is a novel afucosylated human IgG1 mAb that selectively and potently targets human CCR8 with no off-target binding and has the potential to overcome Treg immune suppression in the TME by recruiting T cells
- CHS-114 has demonstrated an acceptable safety profile in heavily pre-treated patients with advanced solid tumors, with no DLTs reported to date
- CHS-114 PK exposure increases with dose, is approximately dose proportional, and the elimination appears linear with a half-life of about 10 days (range 9-17 days)
- Depletion of peripheral CCR8+ Tregs was observed and depletion was maintained over the dosing interval, establishing proof of mechanism
- Preliminary results and acceptable safety profile support further evaluation of CHS-114 in combination treatment with the anti-PD-1 antibody, toripalimab, and other IO agents

Acknowledgments: The authors would like to thank the patients who are participating in this study, their families and caregivers, as well as the investigators and study teams at all clinical sites.