Results from a phase 2 study of triplet blockade of the IL-27, PD-(L)1, and VEGF pathways with casdozokitug (casdozo, CHS-388) in combination with atezolizumab and bevacizumab in patients with unresectable, locally advanced or metastatic hepatocellular carcinoma (uHCC)

Daneng Li,[1] Kun-Ming Rau,[2] Ming-Lung Yu,[3] Hong Jae Chon,[4] Meredith Pelster,[5] Stuart K. Roberts,[6] Oxana Crysler,[7] Gina Vaccaro,[8] Hyung-Don Kim,[9] Koho Iizuka,[10] Hong Tang,[10] Varun N. Kapoor,[10] Daniel J. Chin,[10] Chih-Hung Hsu,[11] Yoon-Koo Kang[9]

[1]City of Hope Comprehensive Cancer Center, Duarte, CA, USA; [2]E-Da Cancer Hospital Kaohsiung City, Taiwan; [3]Kaohsiung Medical University Chung-Ho Memorial Hospital, Kaohsiung Medical University, Kaohsiung City, Taiwan; [4]CHA University – CHA Bundang Medical Center, Gyeonggi-do, South Korea; [5]Sarah Cannon Research Institute, Nashville, TN, USA; [6]The Alfred Hospital, Melbourne, Australia; [7]University of Michigan Health System, Ann Arbor, Michigan, USA; [8]Tennessee Oncology, Nashville, TN, USA; [9]University of Ulsan College of Medicine - Asan Medical Center (AMC), Seoul, South Korea; [10]Coherus BioSciences, Redwood City, CA, USA; [11]National Taiwan University Hospital, Taipei City, Taiwan

Poster presented at ASCO GI 2025

Abstract #605

Copies of this poster obtained through QR codes are for personal use only and may not be reproduced without written permission from ASCO® or the author of the poster.

BACKGROUND

- IL-27 is a member of the IL-12/IL-23 cytokine family comprised of IL-27p28 and EBI3 subunits. It is an immunoregulatory cytokine expressed by myeloid cells, including macrophages and dendritic cells, and dampens T and NK effector function
- IL-27 is highly expressed by tumor-associated macrophages (TAM) in several cancers, including liver (HCC) and lung (NSCLC), and suppresses antitumor immune responses
- Casdozokitug (or casdozo; CHS-388; formerly SRF388) is a first-in-class high affinity human IL-27 antagonistic antibody, which promotes immune activation and stimulates antitumor response
- In a Phase 1 study (NCT04374877), casdozo demonstrated a favorable safety profile and antitumor activity (PR) as a single agent and in combination with PD-1 blockade in indications known to have high levels of IL-27 pathway activation (NSCLC, RCC, and HCC)[1]
- This open-label phase 2 trial examined the potential antitumor activity and safety of casdozo with PD-L1 and VEGF blockade in unresectable/metastatic HCC (NCT05359861). Updated clinical and biomarker data are presented

IL-27 Inhibits T and NK Cell-Driven Antitumor Response in the Tumor Microenvironment

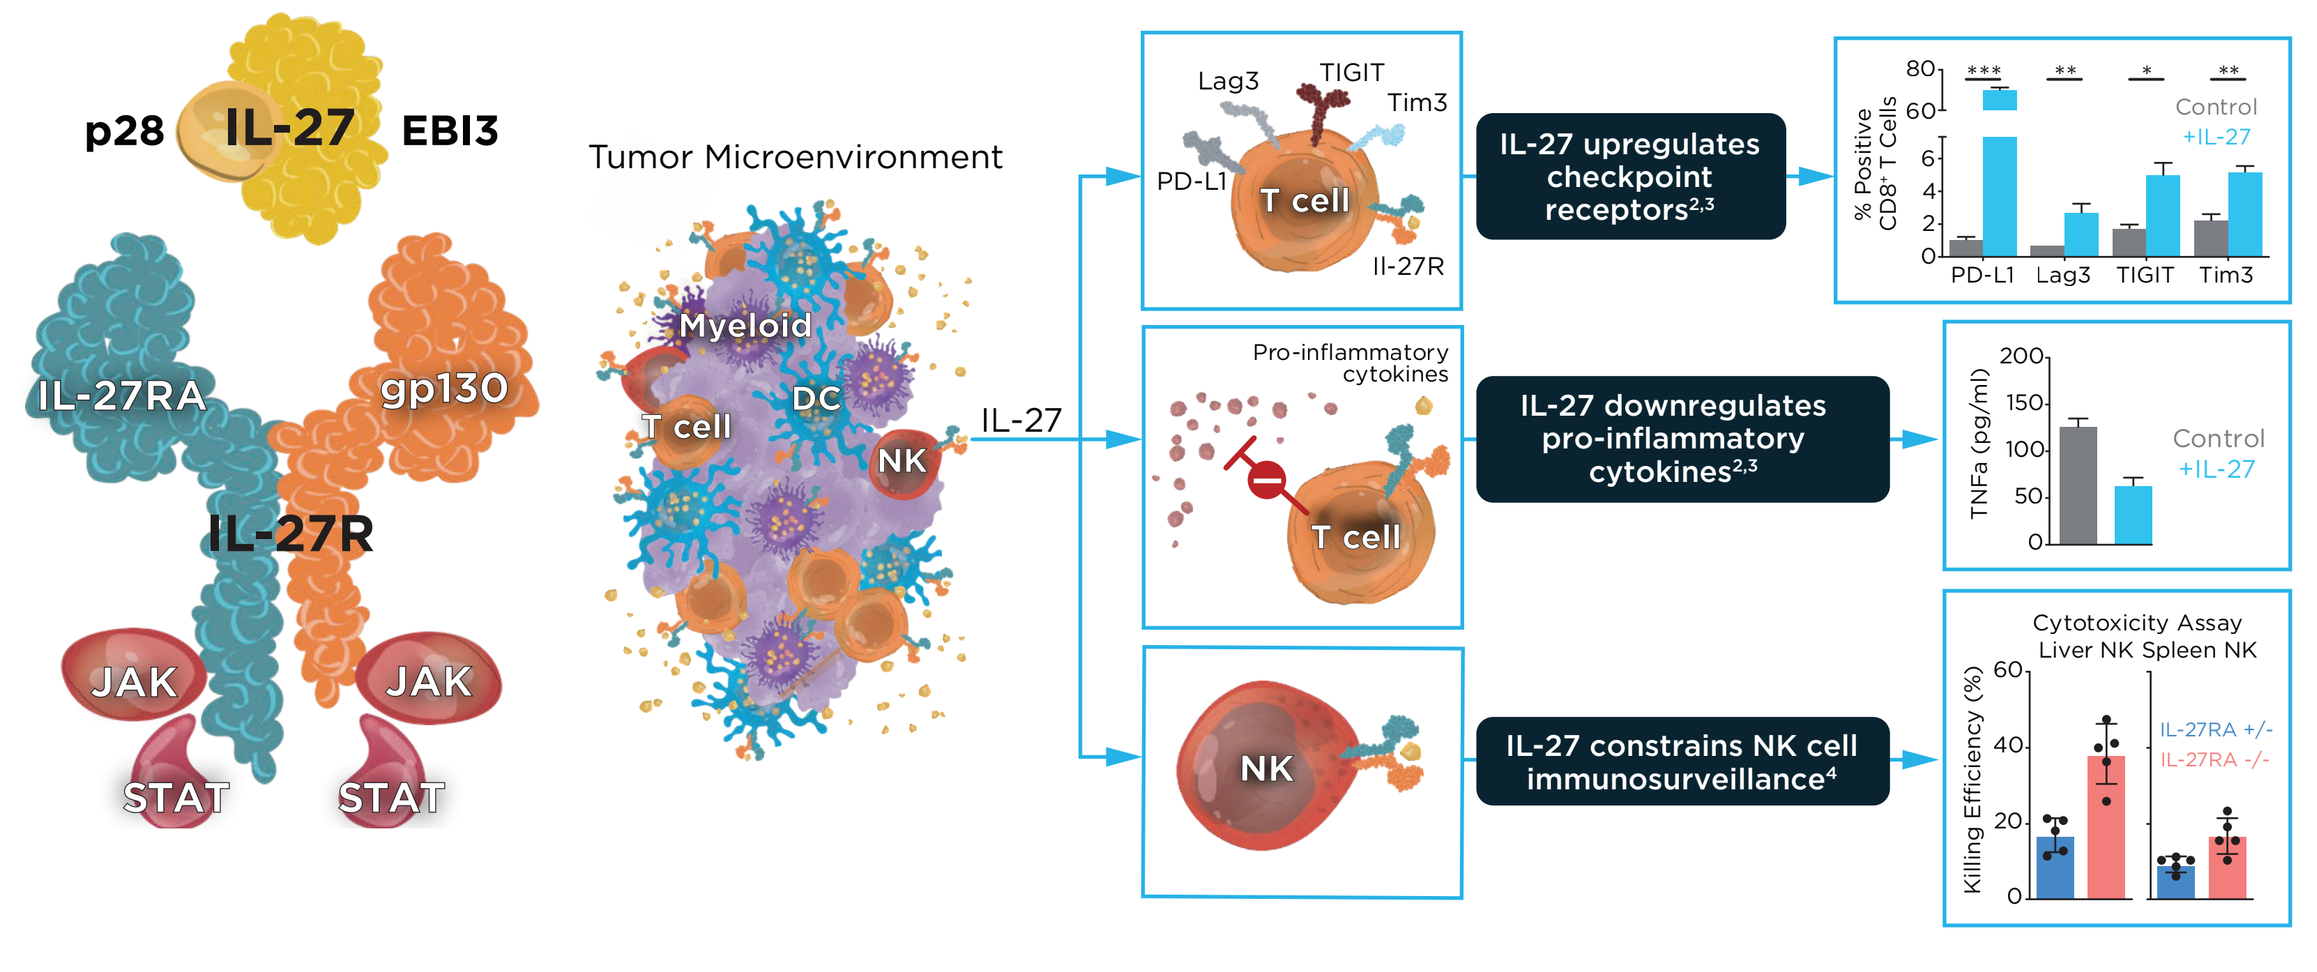

p28
IL-27
EBI3
IL-27RA
gp130
IL-27R
JAK
JAK
STAT
STAT
Tumor Microenvironment
Myeloid
T cell
DC
NK
IL-27
Lag3
TIGIT
Tim3
PD-L1
T cell
Il-27R
IL-27 upregulates checkpoint receptors[2,3]
% Positive CD8+ T Cells
PD-L1 Lag3 TIGIT Tim3
Control
+IL-27
Pro-inflammatory cytokines
T cell
IL-27 downregulates pro-inflammatory cytokines[2,3]
TNFa (pg/ml)
Control
+IL-27
NK
IL-27 constrains NK cell immunosurveillance[4]
Cytotoxicity Assay
Liver NK Spleen NK
Killing Efficiency (%)
IL-27RA +/-
IL-27RA -/-

METHODS

SRF388-201 — Study Schema

SRF388-201: A Phase 2 trial evaluating the efficacy and safety of casdozo in combination with atezolizumab plus bevacizumab in patients with 1L advanced or metastatic HCC

Open-Label Lead-In (N=30)
1L unresectable/metastatic HCC
aPD-(L)1 naïve
≥1 measurable lesion
BCLC B or C
Child-Pugh A
ECOG PS 0-1
Controlled HBV or Cured HCV

→ **Atezolizumab 1200 mg**
Bevacizumab 15 mg/kg
Casdozokitug 10 mg/kg
IV q3w

→
- Primary endpoint: Safety
- Key secondary endpoints: PFS* ORR*, DCR

*RECIST v1.1 (primary) and mRECIST (secondary)

1L, first line; aPD-(L)1, anti-programmed cell death-ligand 1; BCLC, Barcelona Clinic Liver Cancer; DCR, disease control rate; ECOG PS, Eastern Cooperative Oncology Group Performance Status; HBV, hepatitis B virus; HCC, hepatocellular carcinoma; HCV, hepatitis C virus; IV, intravenous; mRECIST, modified Response Evaluation Criteria in Solid Tumors; ORR, overall response rate; PFS, progression-free survival; q3w, every 3 weeks; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1.

RESULTS

Baseline Characteristics

Baseline Characteristics	Casdozo/Atezolizumab/Bevacizumab (n=30)
Median age, years (range)	66 (19, 82)
Sex, n (%)	
Male	23 (76.7)
Female	7 (23.3)
Race, n (%)	
Asian	20 (66.7)
Native Hawaiian or other Pacific Islander	1 (3.3)
White	7 (23.3)
Not reported	2 (6.7)
Region, n (%)	
Asia excluding Japan	18 (60.0)
ROW	12 (40.0)
ECOG PS, n (%)	
0	7 (23.3)
1	23 (76.7)
Stage, n (%)	
Locally advanced	10 (33.3)
Metastatic	20 (66.7)
Macrovascular involvement*, n (%)	7 (23.3)
Varices at study entry, n (%)	3 (10.0)
Child-Pugh Score, n (%)	
A5	27 (90.0)
A6	3 (10.0)
BCLC Stage, n (%)	
B	6 (20.0)
C	24 (80.0)
Viral Status, n (%)	
HBV	16 (53.3)
HCV	5 (16.7)
Uninfected	9 (30.0)
Baseline AFP, n (%)	
<400	16 (53.3)
≥400	14 (46.7)

*Macrovascular involvement = hepatic vein invasion and/or main portal vein invasion.

Time on Therapy (n=30) 1L HCC Fully Enrolled and Data Maturing

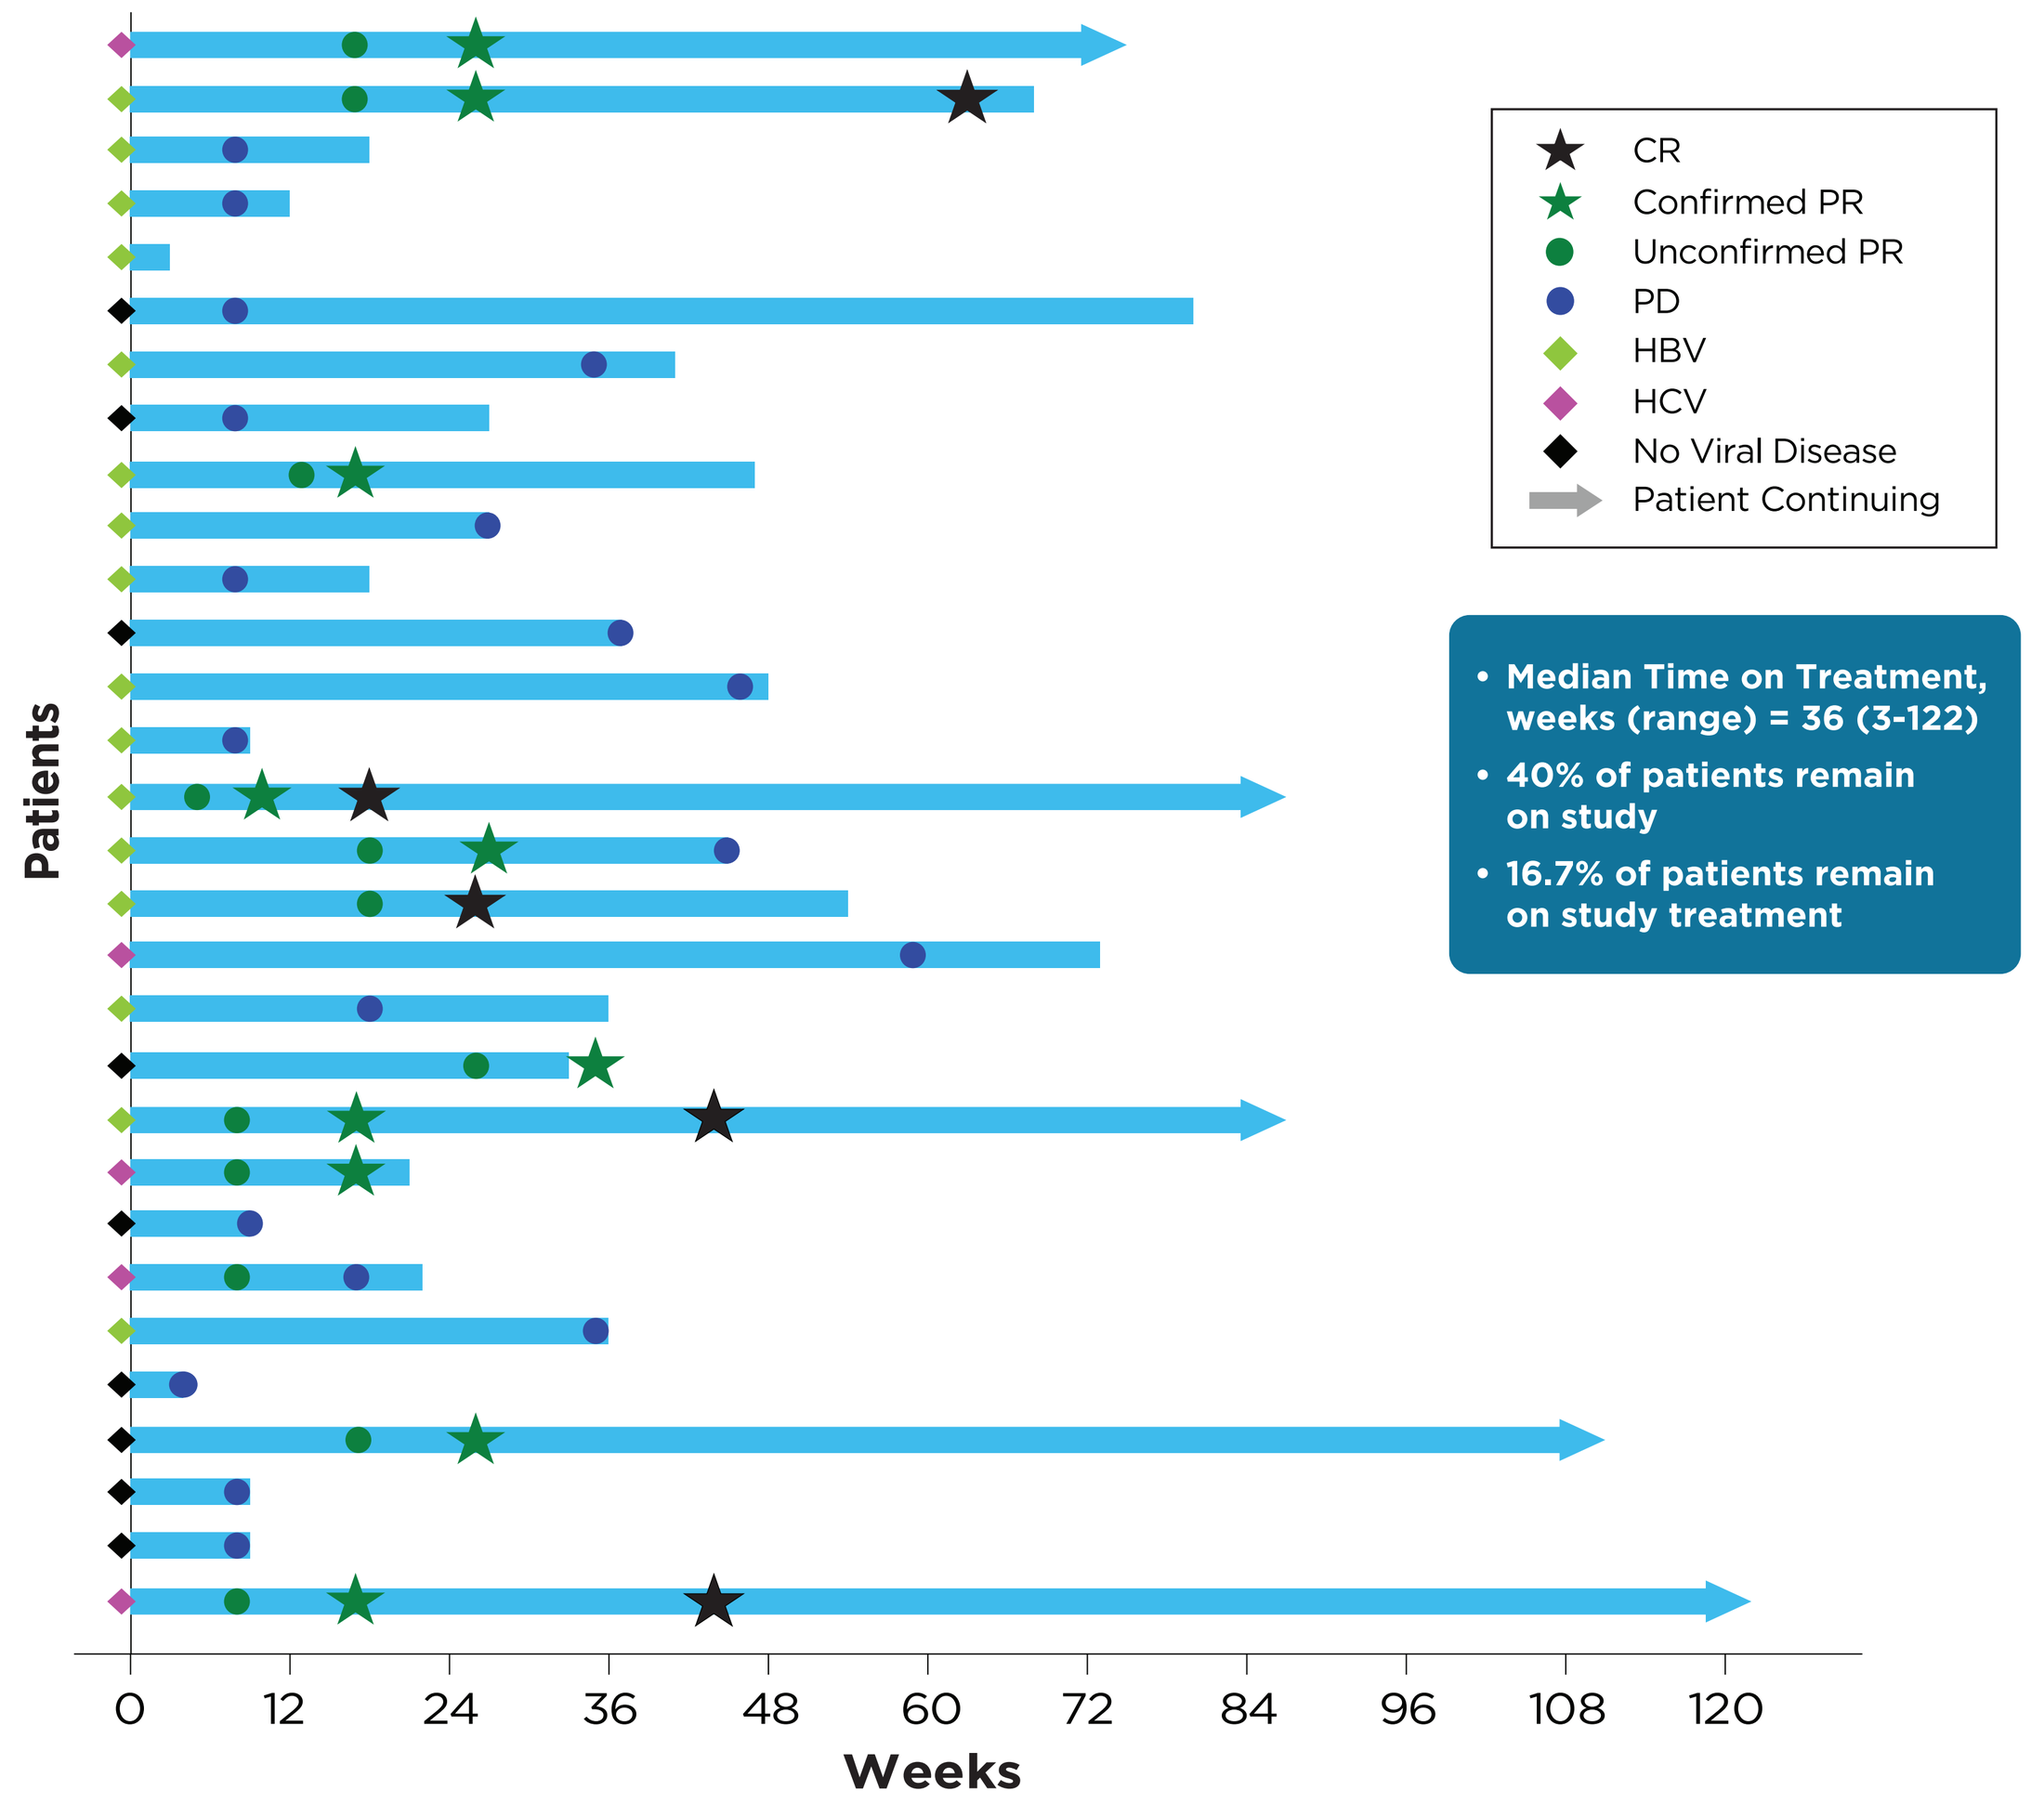

Patients
Weeks
0 12 24 36 48 60 72 84 96 108 120
CR
Confirmed PR
Unconfirmed PR
PD
HBV
HCV
No Viral Disease
Patient Continuing

- Median Time on Treatment, weeks (range) = 36 (3-122)
- 40% of patients remain on study
- 16.7% of patients remain on study treatment

CR, complete response; HBV, hepatitis B virus; HCV, hepatitis C virus; PD, progressive disease; PR, partial response.

Early Activity with Casdozo/Atezolizumab/Bevacizumab

11 Objective Responses per RECIST v1.1 Including 5 CRs

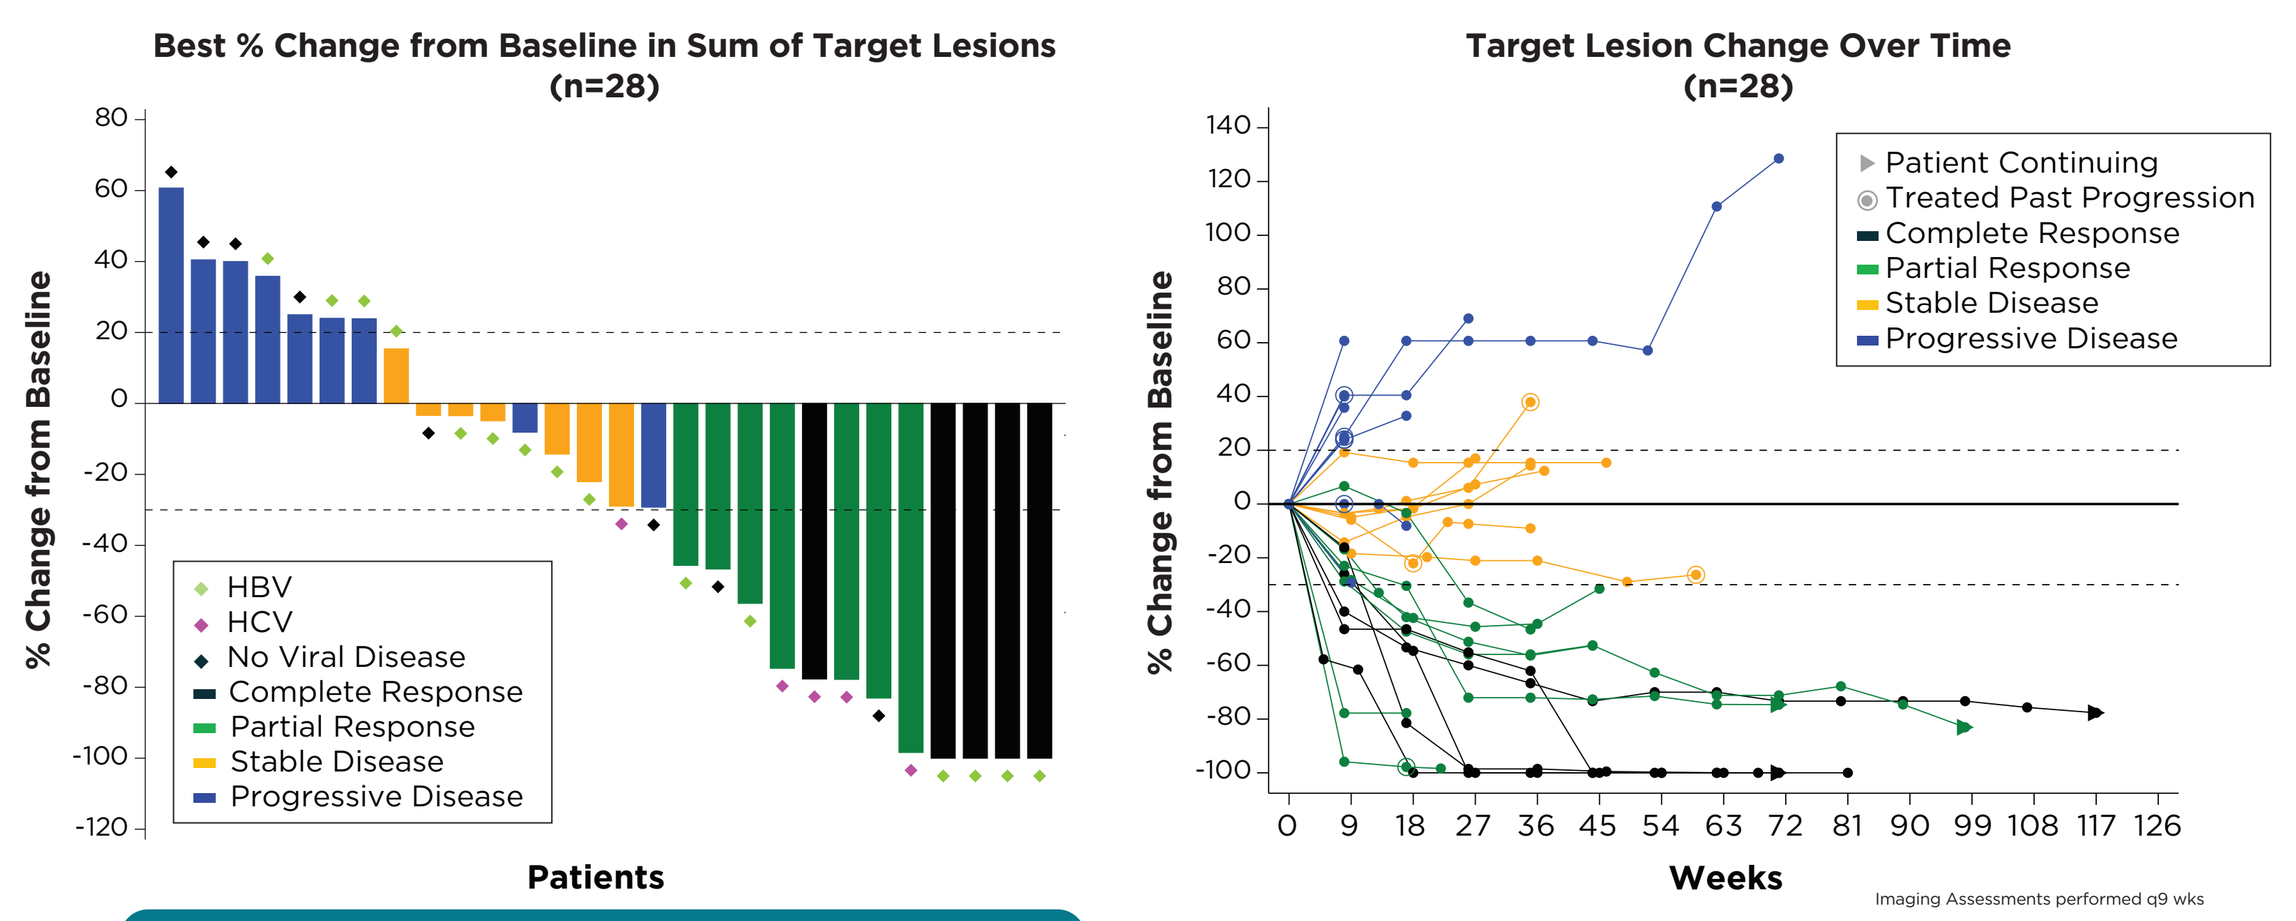

Best % Change from Baseline in Sum of Target Lesions (n=28)
% Change from Baseline
Patients
HBV
HCV
No Viral Disease
Complete Response
Partial Response
Stable Disease
Progressive Disease
Target Lesion Change Over Time (n=28)
Patient Continuing
Treated Past Progression
Weeks
Imaging Assessments performed q9 wks

- >60% of patients with tumor shrinkage on initial scans with 38% ORR to date in response evaluable set: 5 CRs, 6 PRs
- Antitumor activity is observed across viral and nonviral disease subtypes

Best overall response (RECIST v1.1), n (%)	Response evaluable (n=29)
Overall response rate	11 (37.9)
Complete response	5 (17.2)
Partial response (confirmed)	6 (20.7)
Stable disease*	8 (27.6)
Progressive disease	9 (31.0)
Not evaluable	1 (3.4)

* SD includes one patient with an unconfirmed PR

PFS based on RECIST v1.1 Response Evaluable Population

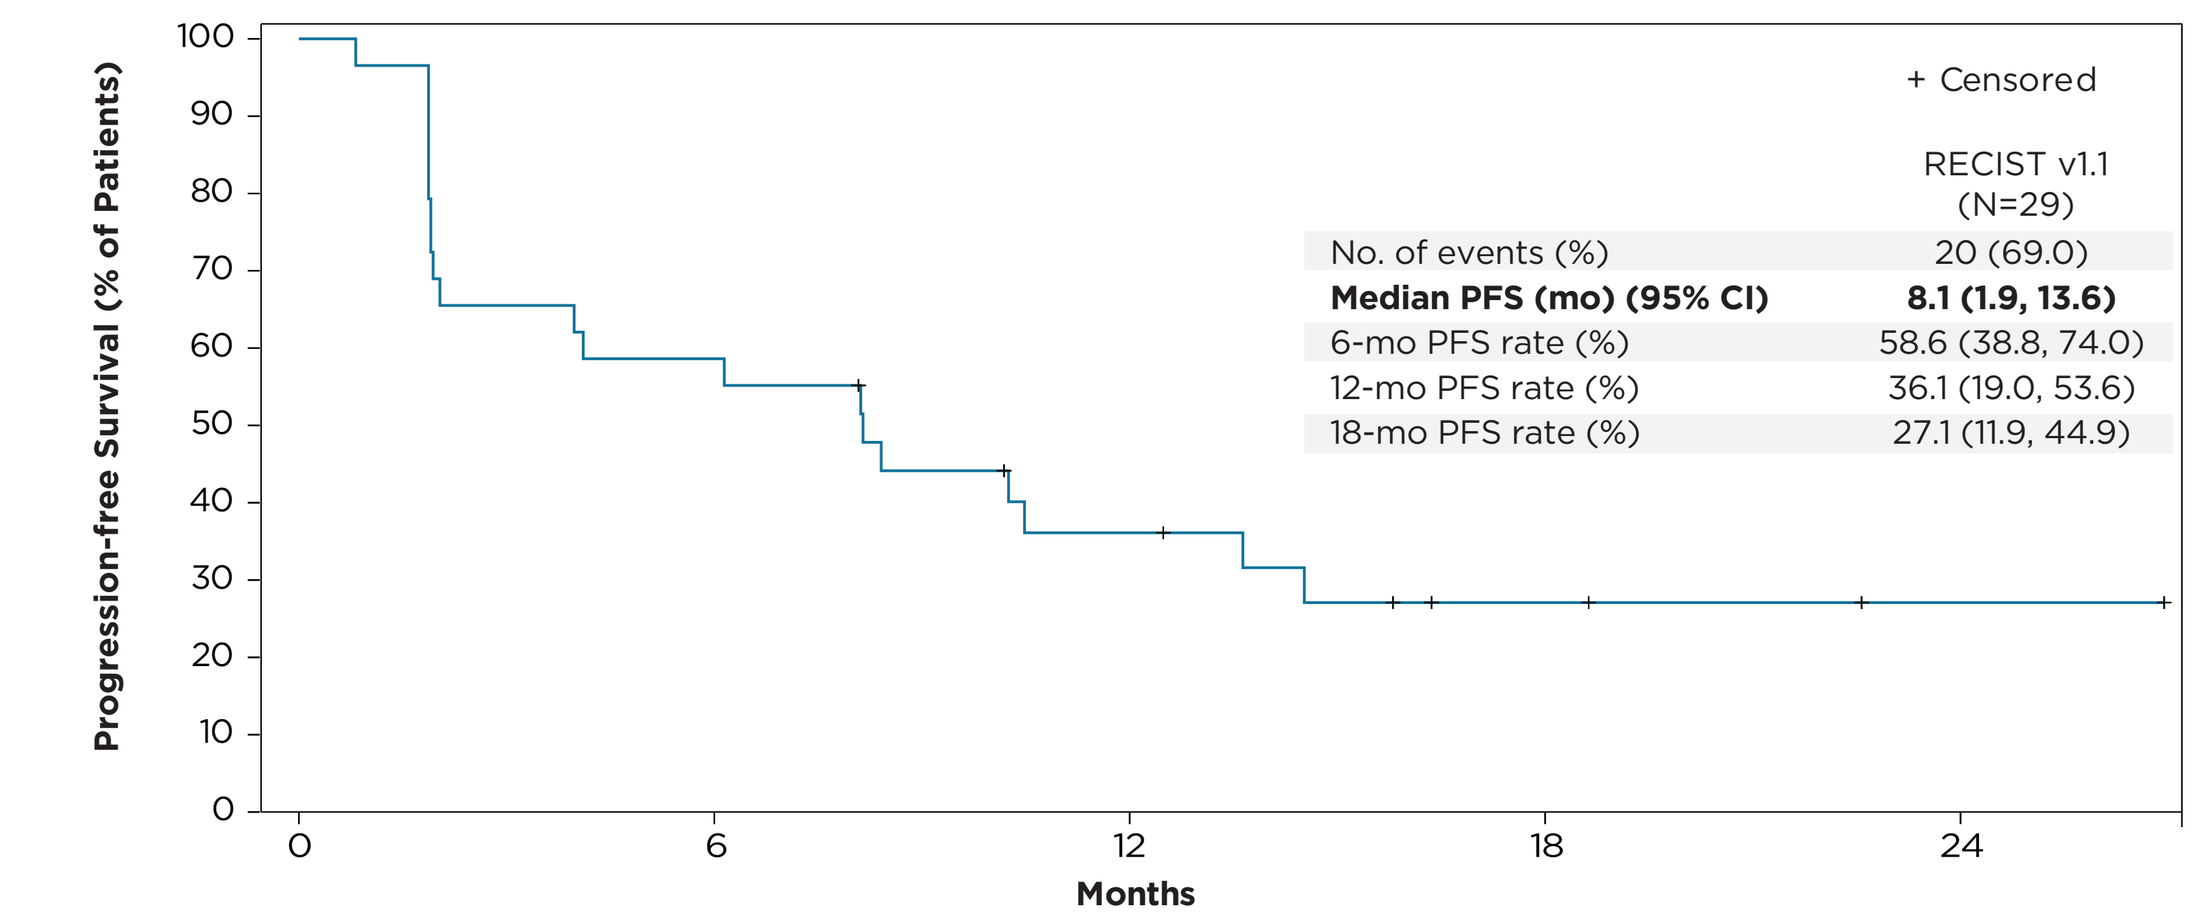

Progression-free Survival (% of Patients)
Months
+ Censored

	RECIST v1.1 (N=29)
No. of events (%)	20 (69.0)
Median PFS (mo) (95% CI)	**8.1 (1.9, 13.6)**
6-mo PFS rate (%)	58.6 (38.8, 74.0)
12-mo PFS rate (%)	36.1 (19.0, 53.6)
18-mo PFS rate (%)	27.1 (11.9, 44.9)

OS

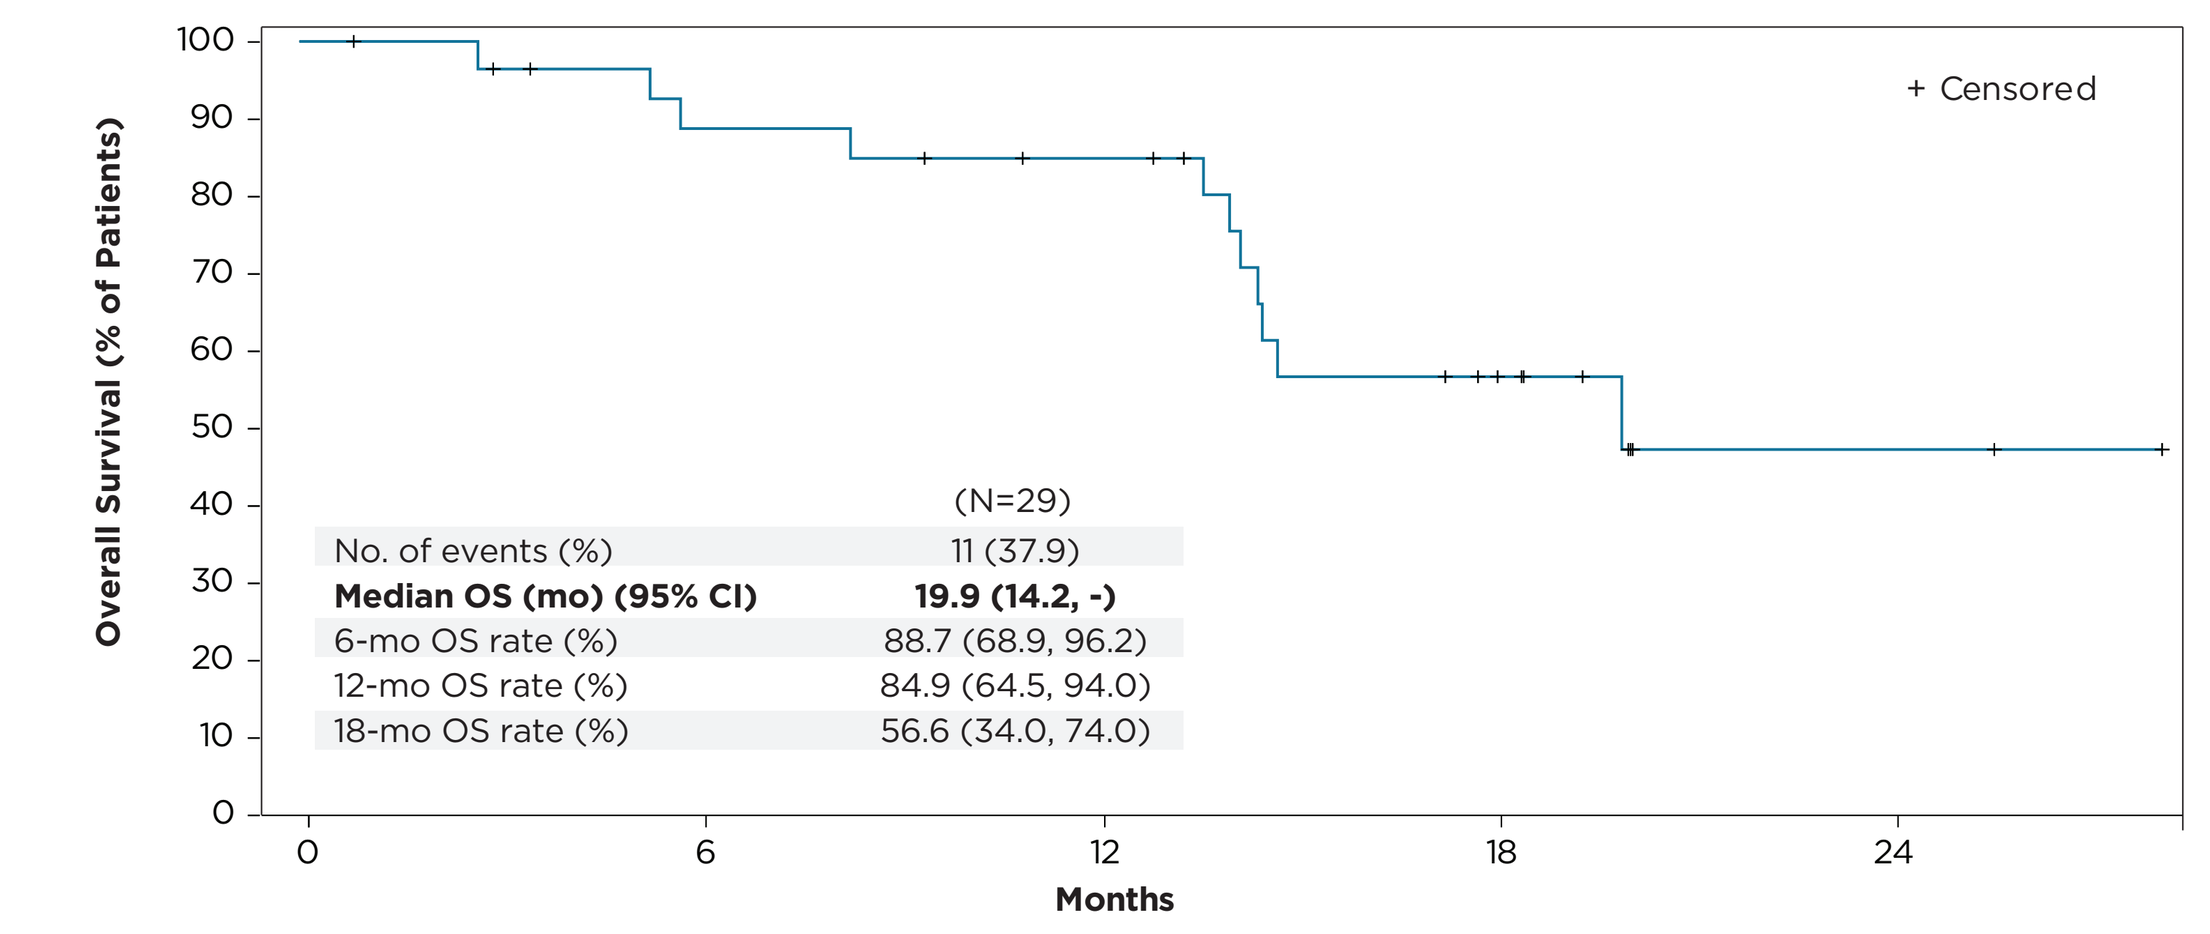

Overall Survival (% of Patients)
Months
+ Censored

	(N=29)
No. of events (%)	11 (37.9)
Median OS (mo) (95% CI)	**19.9 (14.2, -)**
6-mo OS rate (%)	88.7 (68.9, 96.2)
12-mo OS rate (%)	84.9 (64.5, 94.0)
18-mo OS rate (%)	56.6 (34.0, 74.0)

Estimated DCR and DOR in the Response Evaluable Population

	RECIST v1.1 (N=29)	mRECIST (N=28)
DCR, n (%)	17 (58.6)	17 (60.7)
DOR (months)		
Events, n (%)	2 (6.9)	3 (10.7)
Censored, n (%)	9 (31.0)	9 (32.1)
Median (95% CI)	Not reached (6.1, -)	Not reached (12.7, -)
Event-free Rate at, %		
6 months (95% CI)	100.0 (100.0, 100.0)	100.0 (100.0, 100.0)
12 months (95% CI)	90.0 (47.3, 98.5)	90.9 (50.8, 98.7)
18 months (95% CI)	72.0 (23.8, 92.8)	56.8 (14.5, 84.8)

Early mRECIST Activity with Casdozo/Atezolizumab/Bevacizumab

12 Objective Responses Including 5 CRs

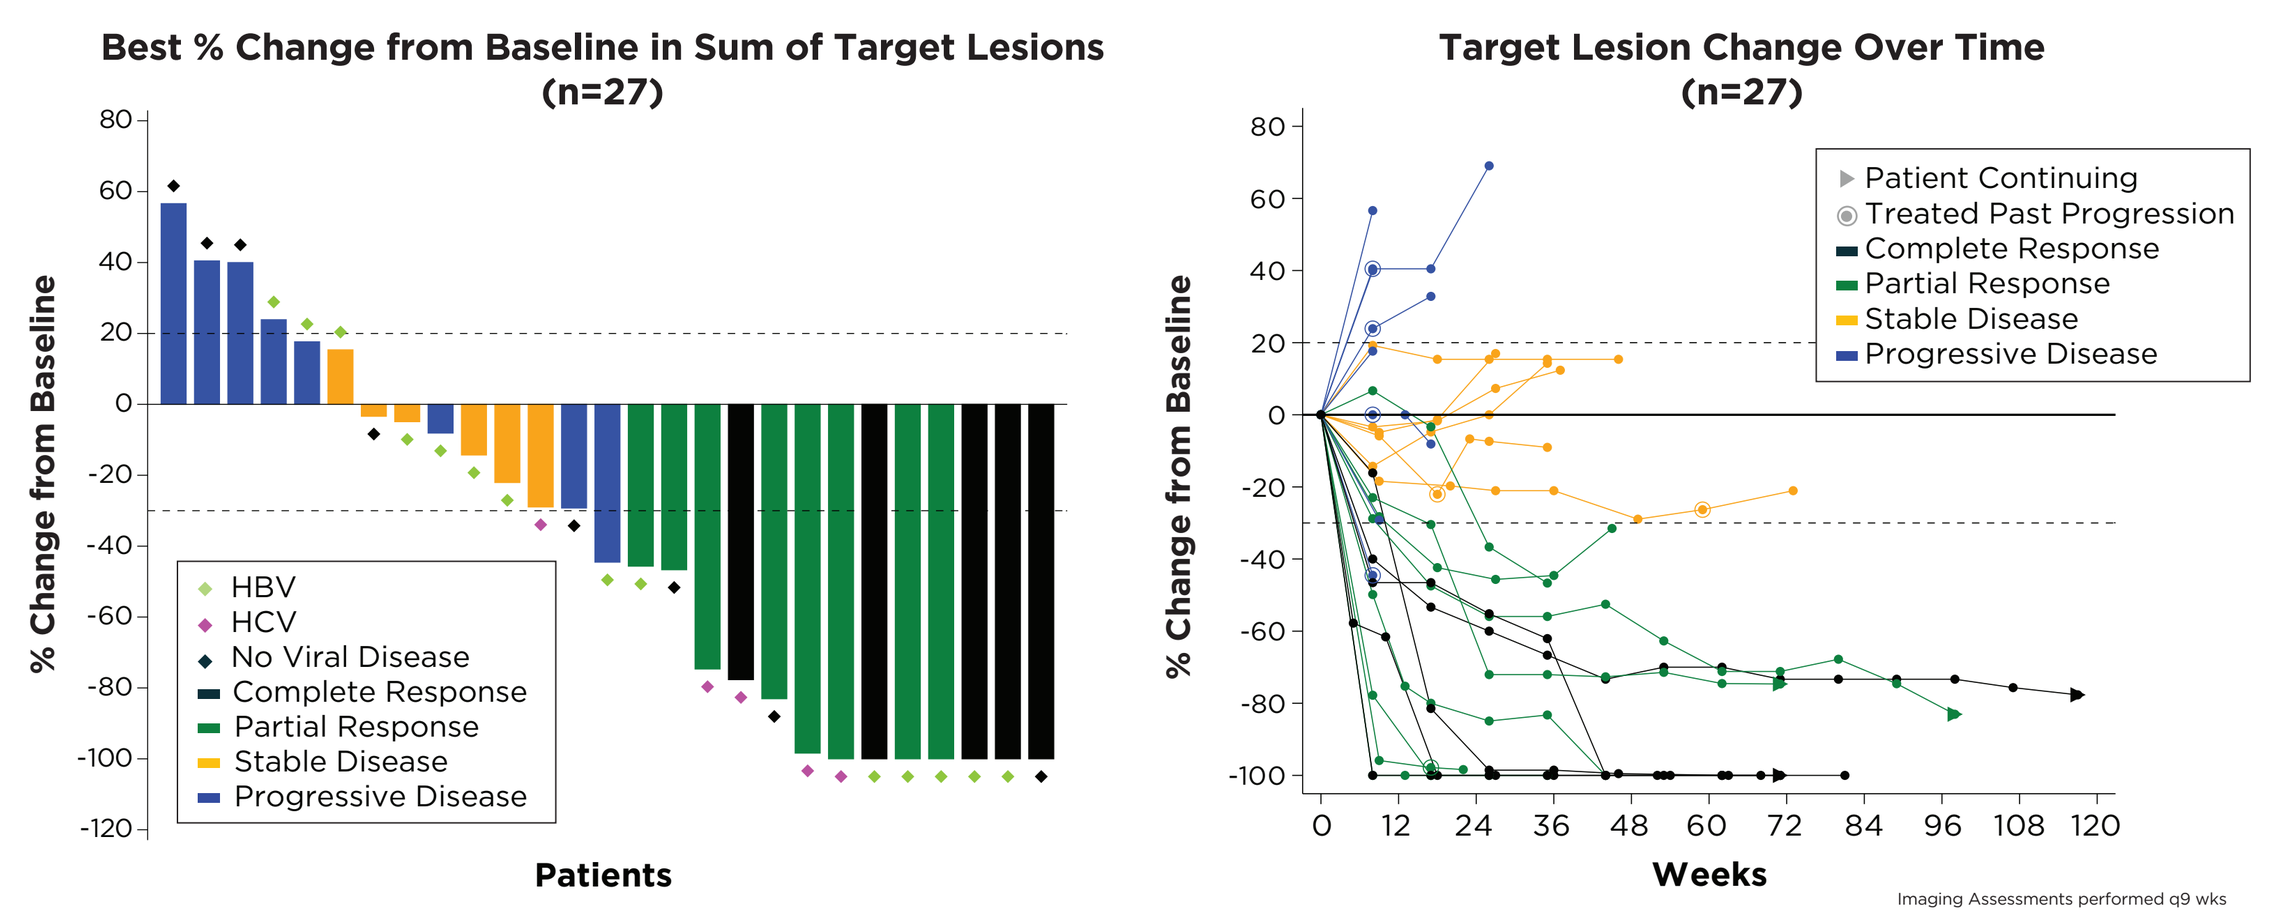

Best % Change from Baseline in Sum of Target Lesions (n=27)
% Change from Baseline
Patients
HBV
HCV
No Viral Disease
Complete Response
Partial Response
Stable Disease
Progressive Disease
Target Lesion Change Over Time (n=27)
Patient Continuing
Treated Past Progression
Weeks
Imaging Assessments performed q9 wks

- Encouraging early data with 43% ORR by mRECIST criteria

Best overall response (mRECIST), n (%)	mRECIST response evaluable (n=28)
Overall response rate	12 (42.9)
Complete response	5 (17.9)
Partial response (confirmed)	7 (25.0)
Stable disease*	7 (25.0)
Progressive disease	8 (28.6)
Not evaluable	1 (3.6)

*SD includes one patient with an unconfirmed PR

Safety Summary of Casdozo/Atezolizumab/Bevacizumab

Overview of Adverse Events	N=30
Treatment emergent adverse event (TEAE), n (%)	30 (100)
Treatment-related AE†, n (%)	27 (90.0)
Grade ≥3 TEAE, n (%)	20 (66.7)
Grade ≥3 treatment-related† AE, n (%)	12 (40.0)
Serious Treatment Emergent Adverse Event (TESAE), n (%)	14 (46.7)
Treatment-related† SAE, n (%)	8 (26.7)
TEAE leading to any study drug discontinuation, n (%)	9 (30.0)
Treatment-related AE leading to any study drug discontinuation, n (%)	7 (23.3)
TEAE leading to CHS-388 discontinuation, n (%)	6 (20.0)
Treatment-related AE leading to CHS-388 discontinuation, n (%)	4 (13.3)
TEAE leading to death, n (%)	3 (10.0)
Treatment-related AE leading to death, n (%)	0

† Related to any treatment in the combination therapy

Casdozo in Combination with SOC (Atezolizumab/Bevacizumab) Is Well Tolerated
Toxicities Consistent with Known AE Profiles of the Study Drugs

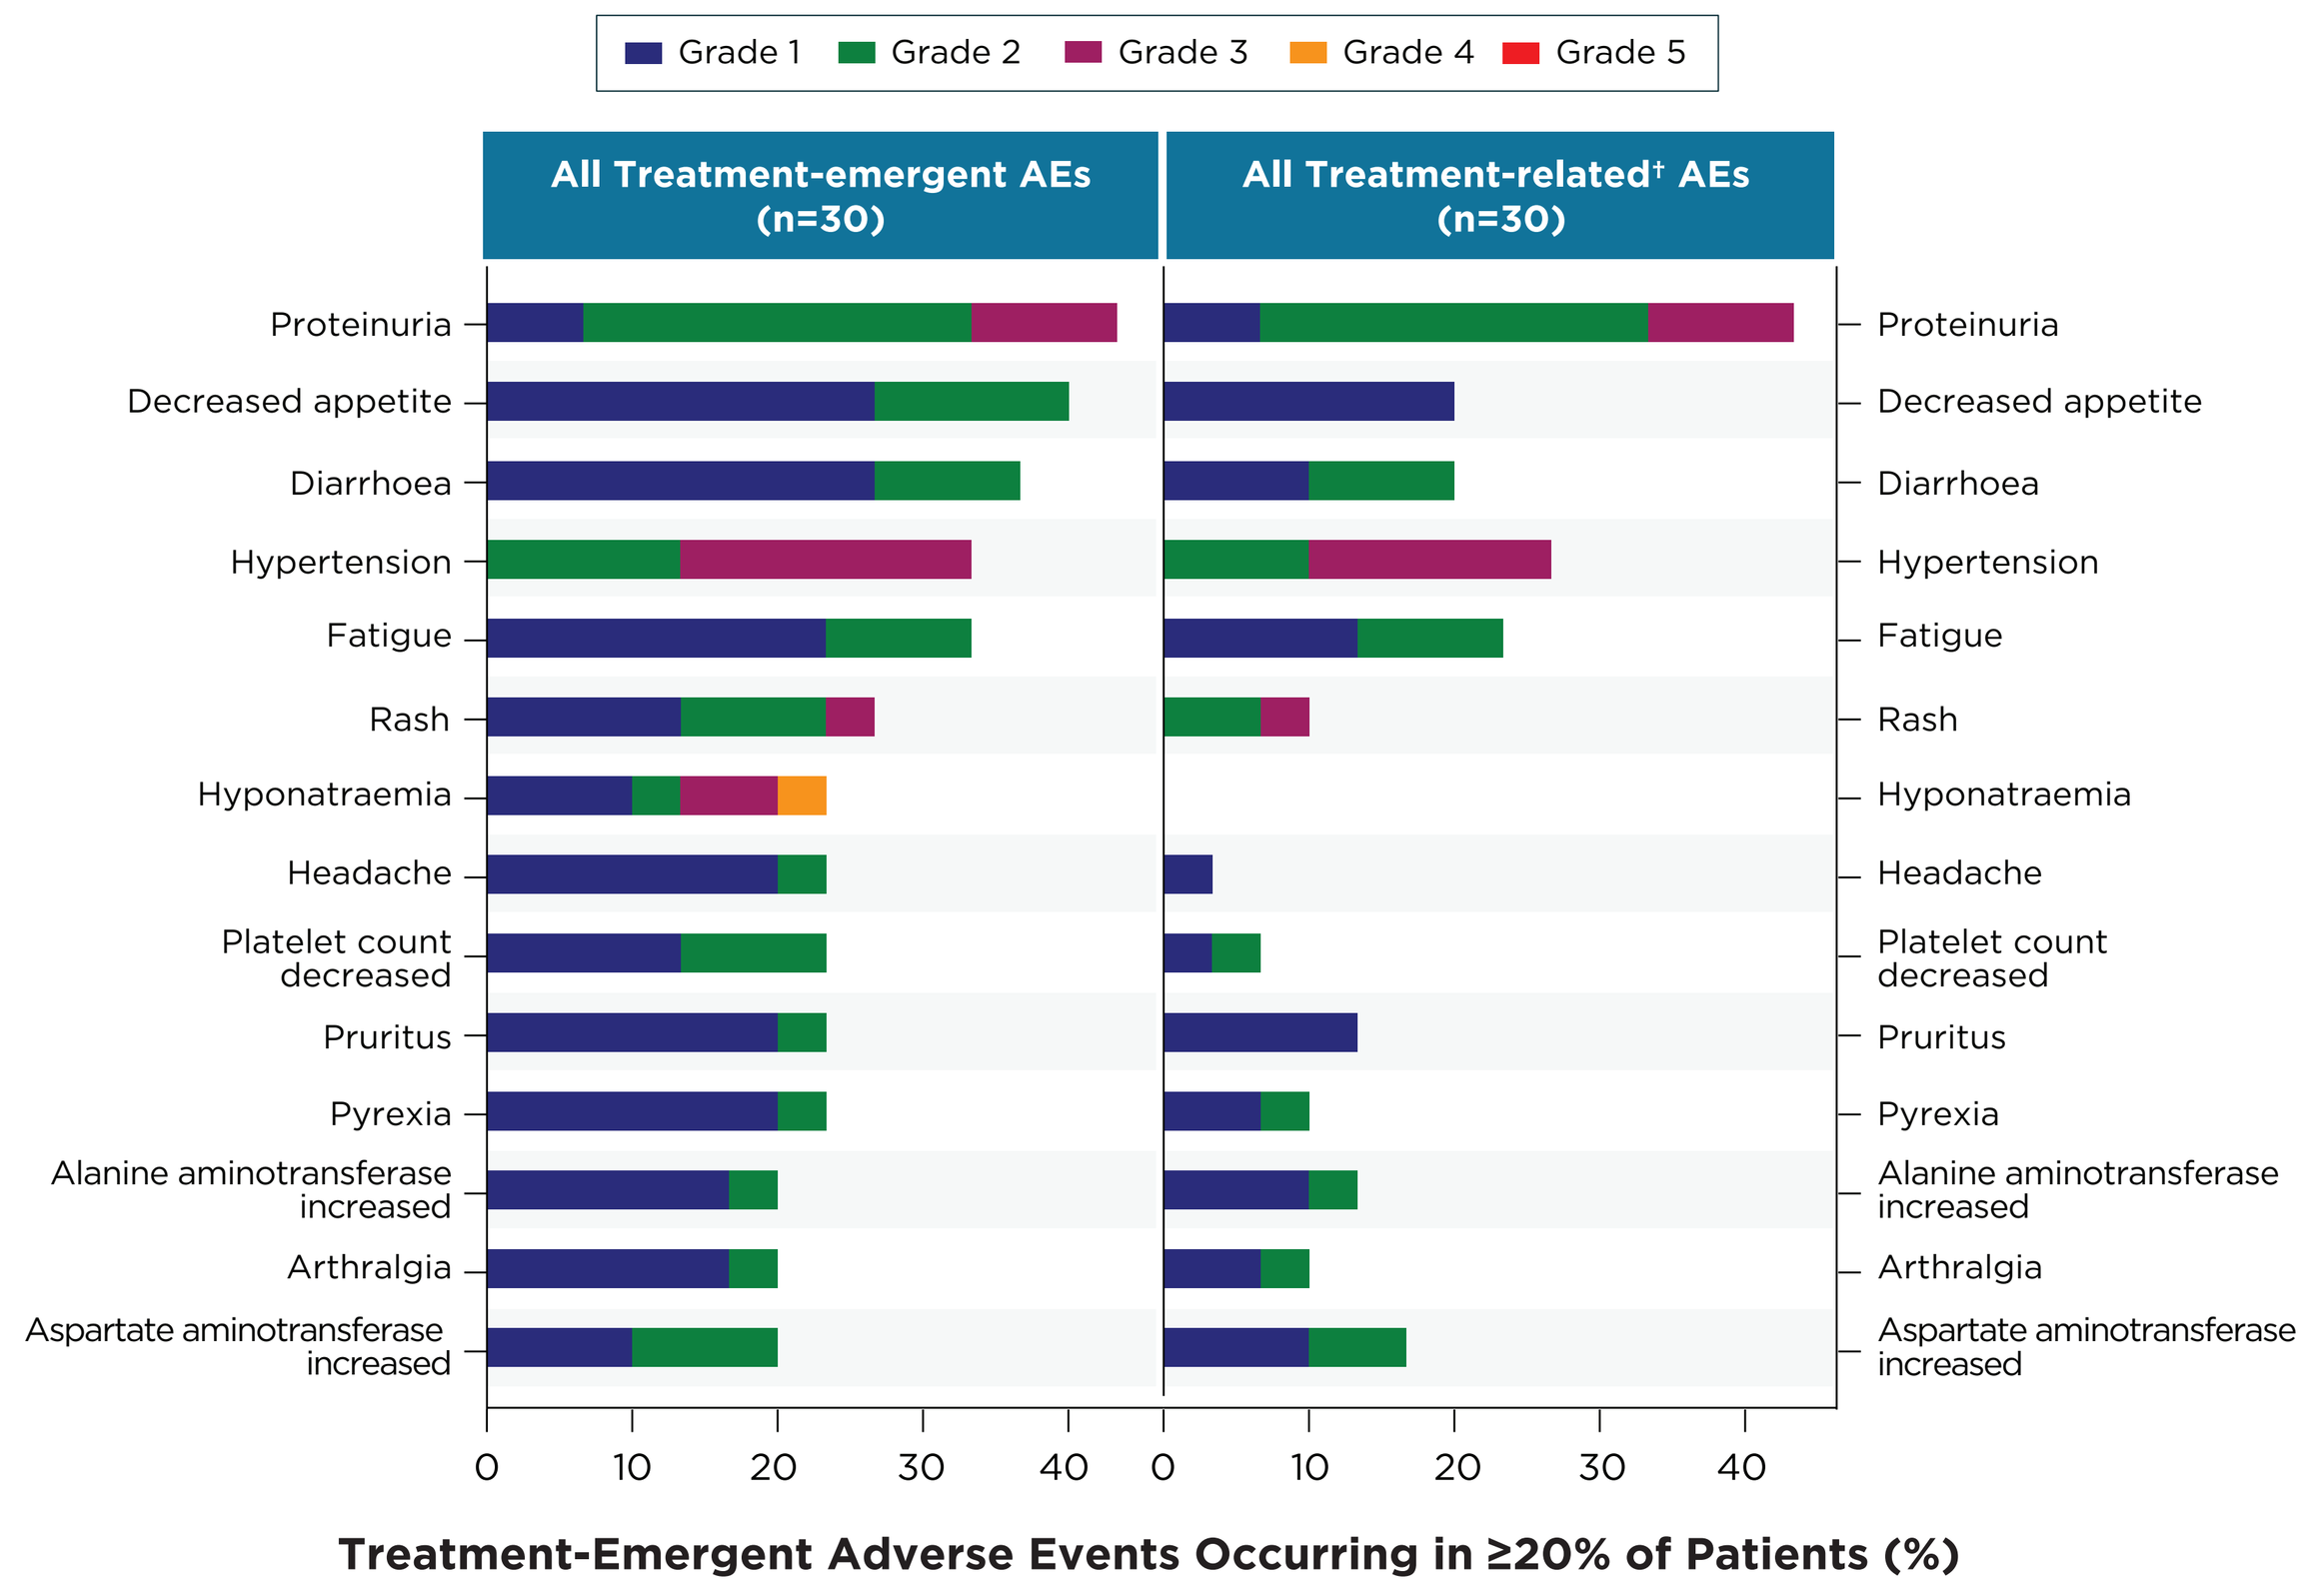

Grade 1
Grade 2
Grade 3
Grade 4
Grade 5
All Treatment-emergent AEs (n=30)
All Treatment-related† AEs (n=30)
Proteinuria
Decreased appetite
Diarrhoea
Hypertension
Fatigue
Rash
Hyponatraemia
Headache
Platelet count decreased
Pruritus
Pyrexia
Alanine aminotransferase increased
Arthralgia
Aspartate aminotransferase increased
0 10 20 30 40
Treatment-Emergent Adverse Events Occurring in ≥20% of Patients (%)

† Related to any treatment in the combination therapy

Preliminary Association of Higher Level of IL-27+ Tumor Associated Macrophages in Archival Tissue Samples with Clinical Response (PR/CR), Small N

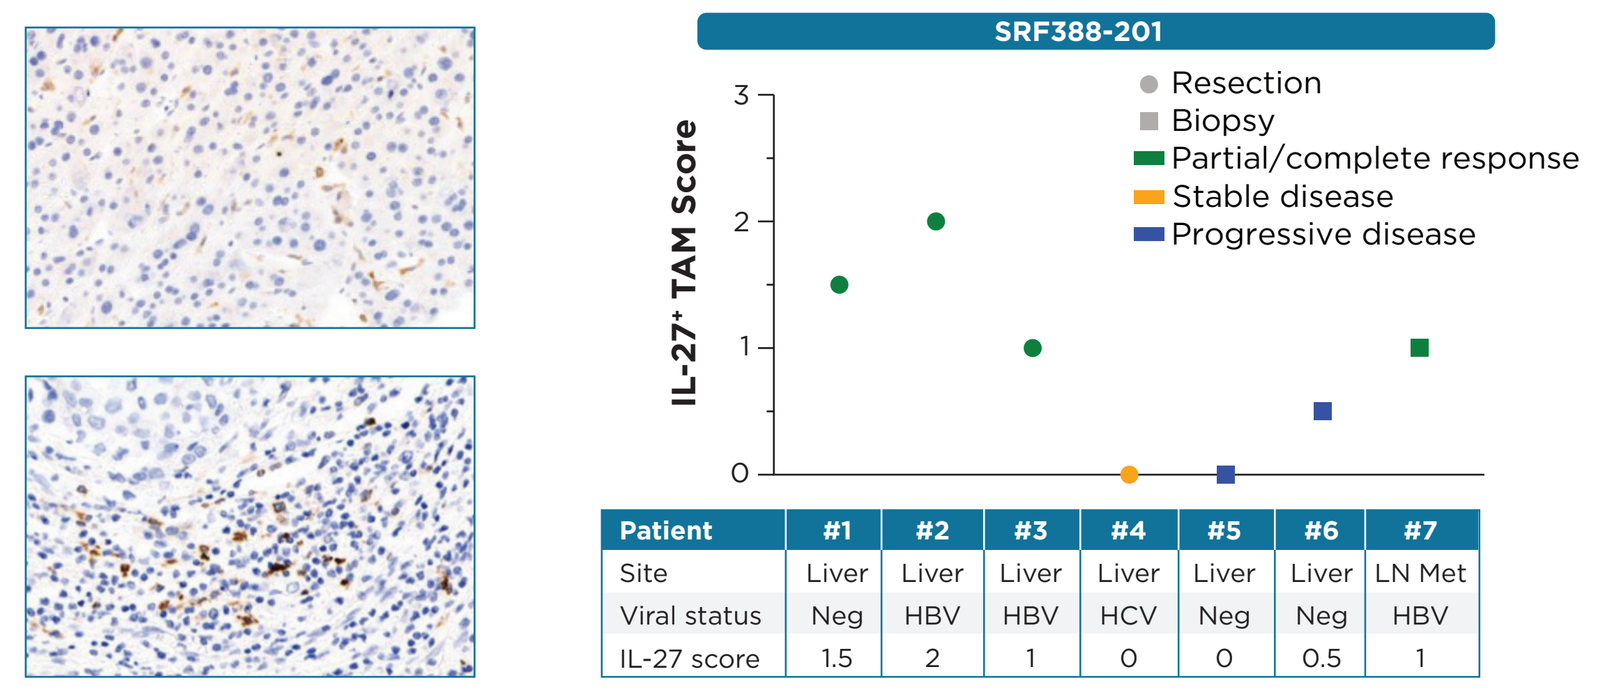

Complete responder (RECIST v1.1 and mRECIST) with HBV+HCC with scattered IL-27+ macrophages with Kupffer cell-like pattern
Complete responder (mRECIST) with non-viral HCC with clusters of IL-27+ TAMs
SRF388-201
IL-27+ TAM Score
Resection
Biopsy
Partial/complete response
Stable disease
Progressive disease

Patient	#1	#2	#3	#4	#5	#6	#7
Site	Liver	Liver	Liver	Liver	Liver	Liver	LN Met
Viral status	Neg	HBV	HBV	HCV	Neg	Neg	HBV
IL-27 score	1.5	2	1	0	0	0.5	1

- Immunohistochemical staining for IL-27 was performed on archival resection and biopsy samples using an independently validated assay
- Samples were scored blindly based on semi-quantitative scoring system (0-3+) evaluating the abundance of IL-27+ tumor associated macrophages (TAM)

Pharmacodynamics and Casdozo/Atezolizumab/Bevacizumab Response Associated Signatures in Patients with HCC

Casdozo/Atezolizumab/Bevacizumab Pharmacodynamic Activity on Immune Activation (serum proteomics)

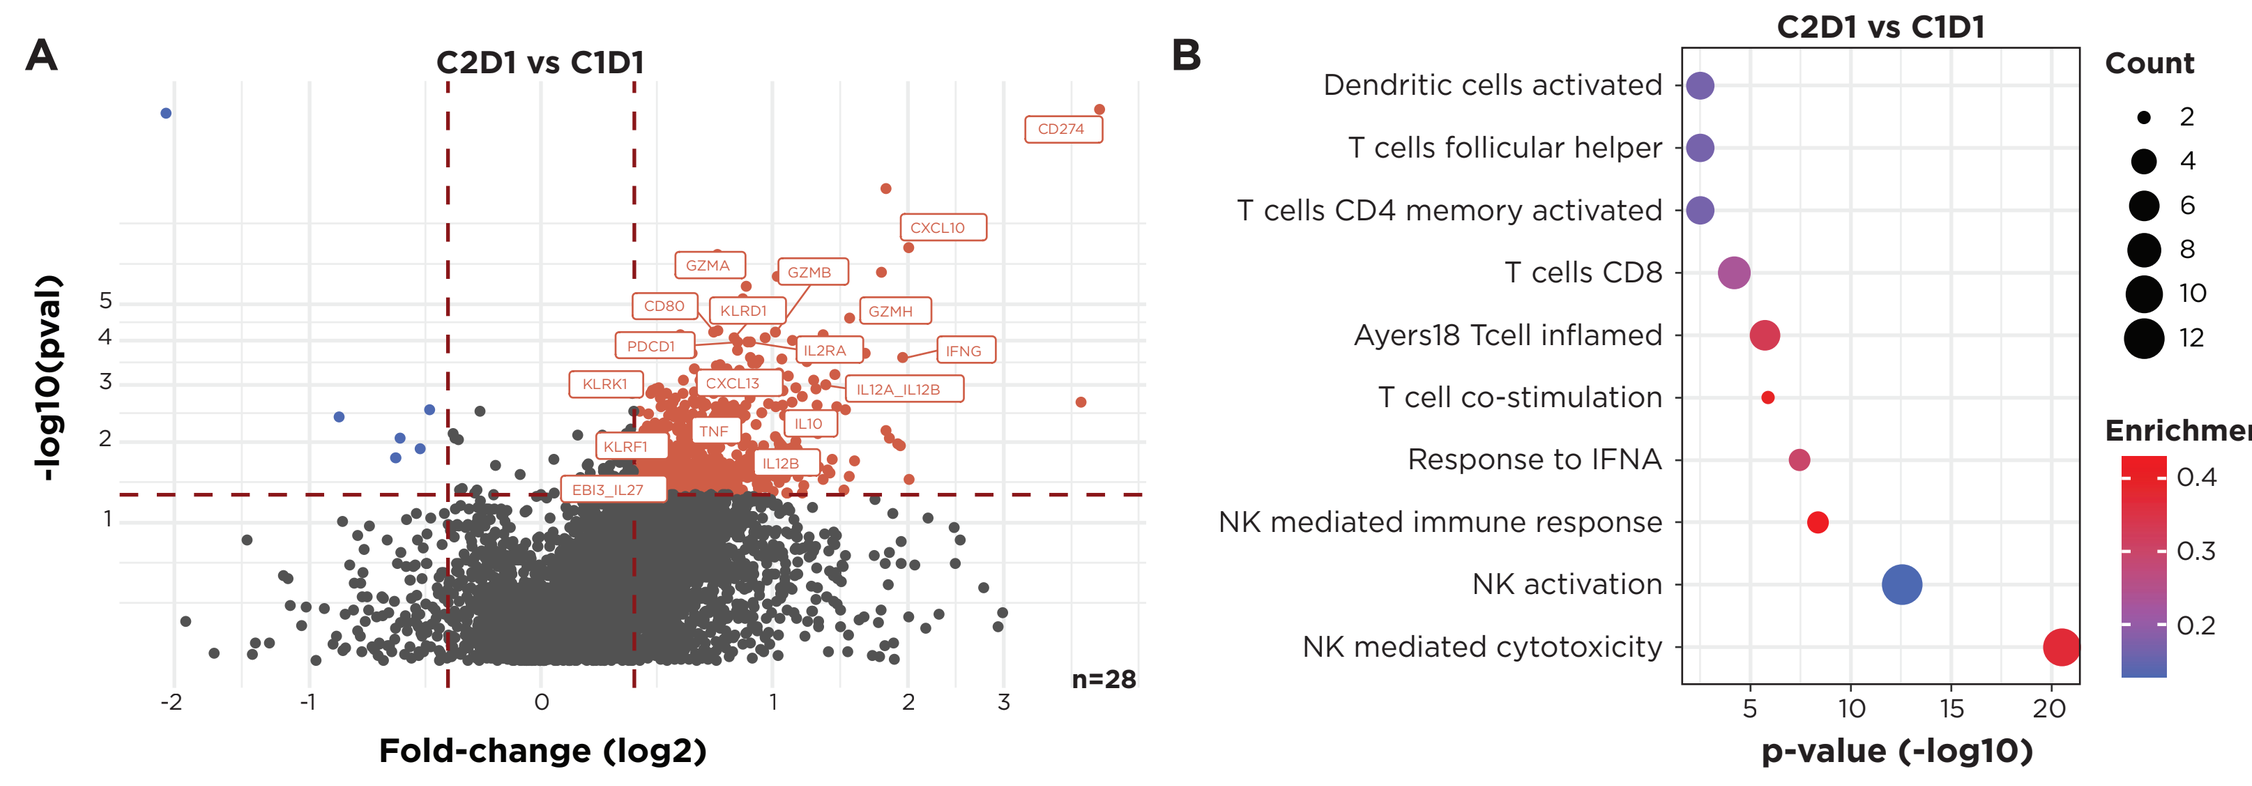

A
C2D1 vs C1D1
-log10(pval)
Fold-change (log2)
n=28
CD274
CXCL10
GZMA
GZMB
CD80
KLRD1
GZMH
PDCD1
IL2RA
IFNG
KLRK1
CXCL13
IL12A_IL12B
TNF
IL10
KLRF1
IL12B
EBI3_IL27
B
C2D1 vs C1D1
Dendritic cells activated
T cells follicular helper
T cells CD4 memory activated
T cells CD8
Ayers18 Tcell inflamed
T cell co-stimulation
Response to IFNA
NK mediated immune response
NK activation
NK mediated cytotoxicity
p-value (-log10)
Count
Enrichment

C Responding (R) vs Non-Responding (NR) Patients Show on Treatment IL-27 Signaling Inhibition and Immune Activation

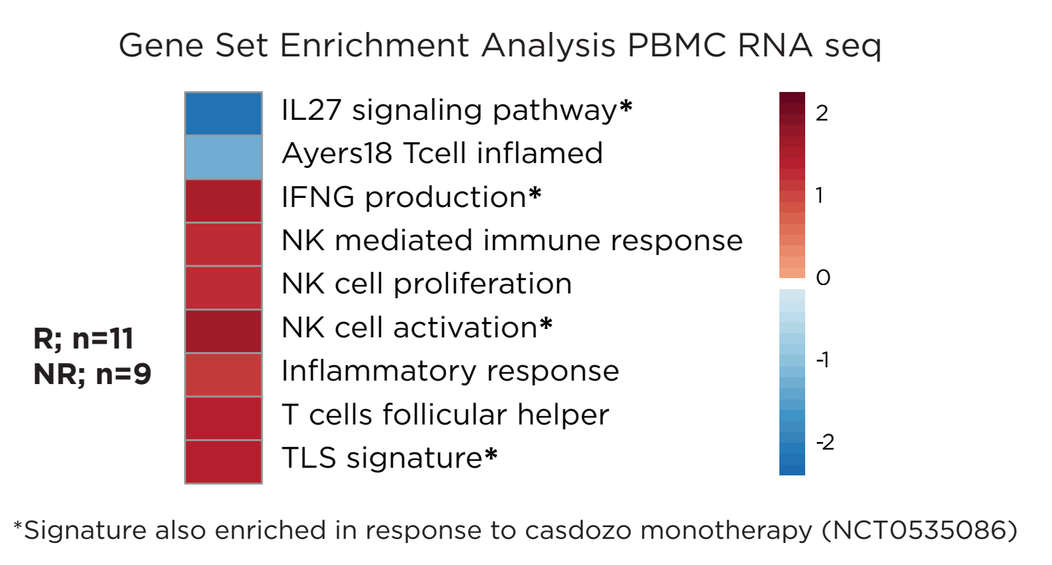

Gene Set Enrichment Analysis PBMC RNA seq
R; n=11
NR; n=9
IL27 signaling pathway*
Ayers18 Tcell inflamed
IFNG production*
NK mediated immune response
NK cell proliferation
NK cell activation*
Inflammatory response
T cells follicular helper
TLS signature*

*Signature also enriched in response to casdozo monotherapy (NCT0535086)

Casdozokitug in combination with atezolizumab and bevacizumab promotes NK and T cell activation and inhibits IL-27 signaling. Serum samples were analyzed with Olink Explore HT platform. Several cytolytic, NK and Th1 related immune activation proteins and pathways were induced in response to treatment. **(A)** Volcano plot of select upregulated (red) and downregulated (blue) proteins (Cycle 2 day 1 [C2D1] vs Cycle 1 day 1 [pretreatment]; n=28). Dashed horizontal and vertical lines represent p < 0.05 and fold change (FC) > 1.25. **(B)** Pathway analysis of serum proteins that were significantly upregulated by treatment using the Fisher's exact t-test of GOBP and LM22 derived genesets.[5,6,7] **(C)** GSEA of paired PBMC RNAseq samples of responders (R; CR.PR, n=11) vs non-responders (NR; PD, n= 9) after treatment (C2D1 vs pretreatment). Response associated increase in NK and T cell activation pathway and inhibition of IL-27 pathway was observed. Pathways upregulated in response to casdozo alone treatment highlighted (NCT0535086). The heatmap shows the normalized enrichment scores from a combination of GOBP, internal genesets and LM22 derived genesets.[5,6,7]

CONCLUSIONS

- Triplet blockade of IL-27, PD-(L)1, and VEGF pathways with casdozo/atezolizumab/bevacizumab continues to show a manageable safety profile with promising antitumor activity in HCC that warrants continued exploration
- Casdozo in combination with atezolizumab/bevacizumab demonstrated a CR rate of 17.2% which is higher than the 3-8% CR rate reported in prior Phase 3 studies in HCC (IMbrave150 and HIMALAYA).[8,9,10] Casdozo triplet treatment resulted in an ORR of 37.9% and a median PFS of 8.1 months achieving the efficacy thresholds for further evaluation
- Toxicity was consistent with the known profiles of atezolizumab and bevacizumab, with no new safety signals identified
- Casdozo/atezolizumab/bevacizumab treatment resulted in IL-27 signaling inhibition and immune activation in first line HCC patients in responders vs nonresponders. Immune activation biomarkers were consistent with casdozo single agent treatment
- A randomized, controlled Phase 2 study (NCT06679985) is currently underway to evaluate the efficacy of casdozo in combination with toripalimab and bevacizumab compared to toripalimab/bevacizumab alone. The toripalimab/bevacizumab combination previously demonstrated superiority over sorafenib in the recent phase 3 HEPATORCH trial[11]

REFERENCES:
1. Marron TU, et al. *Ann Oncol.* 2023;20(suppl_1): 100589-100589. 10.1016/iotech/iotech100589. **2.** Chihara N, et al. *Nature.* 2018;558(7710): 454-459. **3.** DeLong JH, et al. *Immunohorizons.* 2019;3(1):13-25. **4.** Aghayev T, et al. *Cancer Discov.* 2022;12(8):1960-1983. **5.** Chen B, et al. *Methods Mol Biol.* 2018;1711:243-259. **6.** Meylan M, et al. *Immunity*. 2022;55(3):527-541. **7.** Hill J, et al. *J ImmunoTherap Cancer.* 2021;9:doi: 10.1136/jitc-2021-SITC2021.674. **8.** Finn RS, et al. *N Engl J Med.* 2020;382:1894-1905. **9.** Abou-Alfa GK, et al. *NEJM Evid.* 2022;1(8):EVIDoa2100070. **10.** Cheng AL, et al. *J. Hepatol.* 2022;76: 862–873. **11.** Yinghong S, et al. Oral presentation at: CSCO 2024; September 27, 2024; Xiamen, China.